Immunogenicity Analysis of Chinese Hamster Ovary (CHO) Host Cell Protein Contaminants in Therapeutic Protein Formulations

Andres H. Gutiérrez[1], Lenny Moise[1,2], Frances Terry[2], Chris Bailey-Kellogg[3], William D. Martin[2], Anne S. De Groot[1,2]

[1]Institute for Immunology and Informatics, University of Rhode Island, Providence, RI; [2]EpiVax, Inc., Providence, RI; [3]Dartmouth College, Hanover, NH

Abstract

Concerns over immune responses to contaminating host cell proteins (HCPs) in therapeutics have recently emerged. For example, the presence of Chinese hamster ovary (CHO)-derived HCPs contributed to the cancellation of two phase III clinical trials in 2012 for Inspiration's IB1001, a recombinant factor IX produced in CHO cells. These trials were cancelled due to the development of anti-CHO antibodies at higher levels than expected in patients treated with a protein drug. Publication of the CHO-K1 genome and transcriptome offers an opportunity to analyze and understand the immunogenic potential of CHO HCPs.

We analyzed the CHO-K1 genome, the CHO-K1 transcriptome, published CHO proteins identified in biotherapeutics, and mouse secreted proteins downloaded from the LOCATE and UniProtKB/Swiss-Prot databases. These datasets were screened for potential CD4+ T cell immunogenicity using the epitope-mapping algorithm EpiMatrix and overall protein immunogenicity scores calculated. Human homologs discovered by BLAST were similarly screened for T-cell epitope content; predicted immunogenicity was then adjusted for human homology.

Further study is required to assess the potential for autoimmune responses, as several widely publicized cases of severe adverse immune responses to autologous therapeutic proteins are published and the FDA and EMA take the potential for such effects very seriously. Finally, we built a website (CHOPPI) to give protein manufacturers free access to information on potential immunogenicity of CHO-K1 sequences. We look forward to its further development in collaboration with protein therapeutics industry, as this and additional means of performing immunogenicity analysis continue to be refined.

CHO Cell Protein

- CHO-derived Host Cell Protein (HCP) contaminants in biotherapeutics can potentially generate immune responses.
- Of significant concern are immune responses that may be cross-reactive with human T cell epitopes.

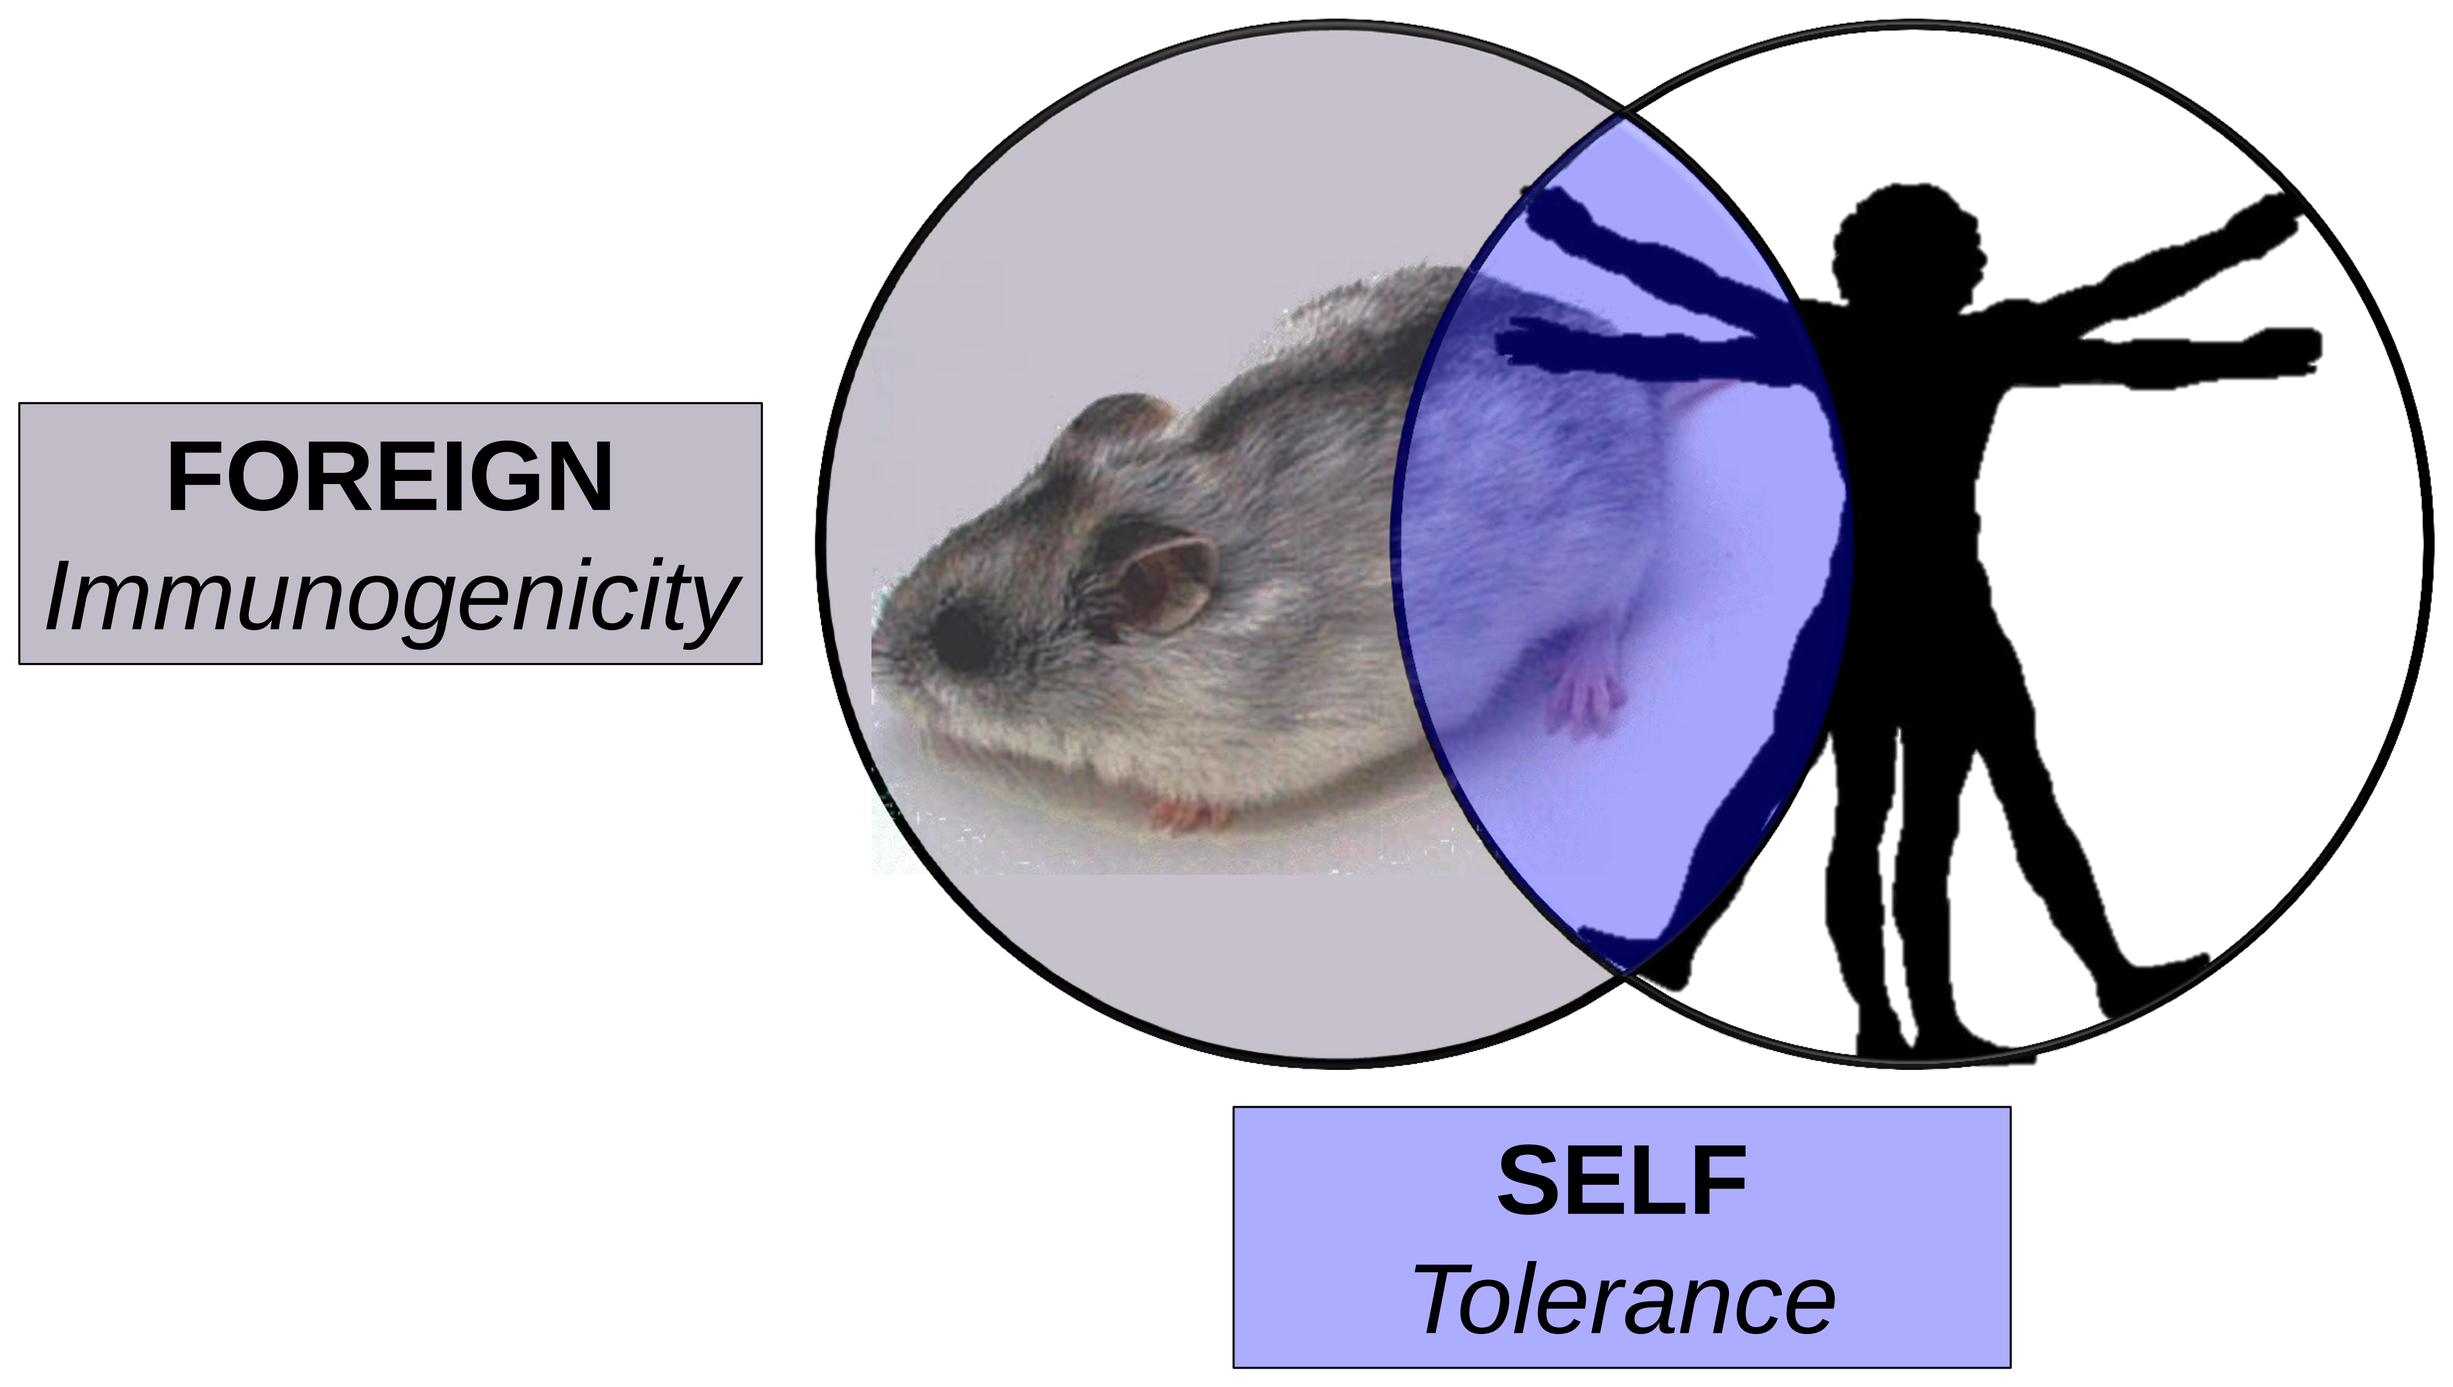

FOREIGN
Immunogenicity
SELF
Tolerance

Figure 1. Immune response to mammalian proteins.

- The availability of the CHO genome and transcriptome has made it possible to apply immunoinformatics tools to HCP analysis.
- This significantly accelerates research on HCP immunogenicity and cross-reactivity.

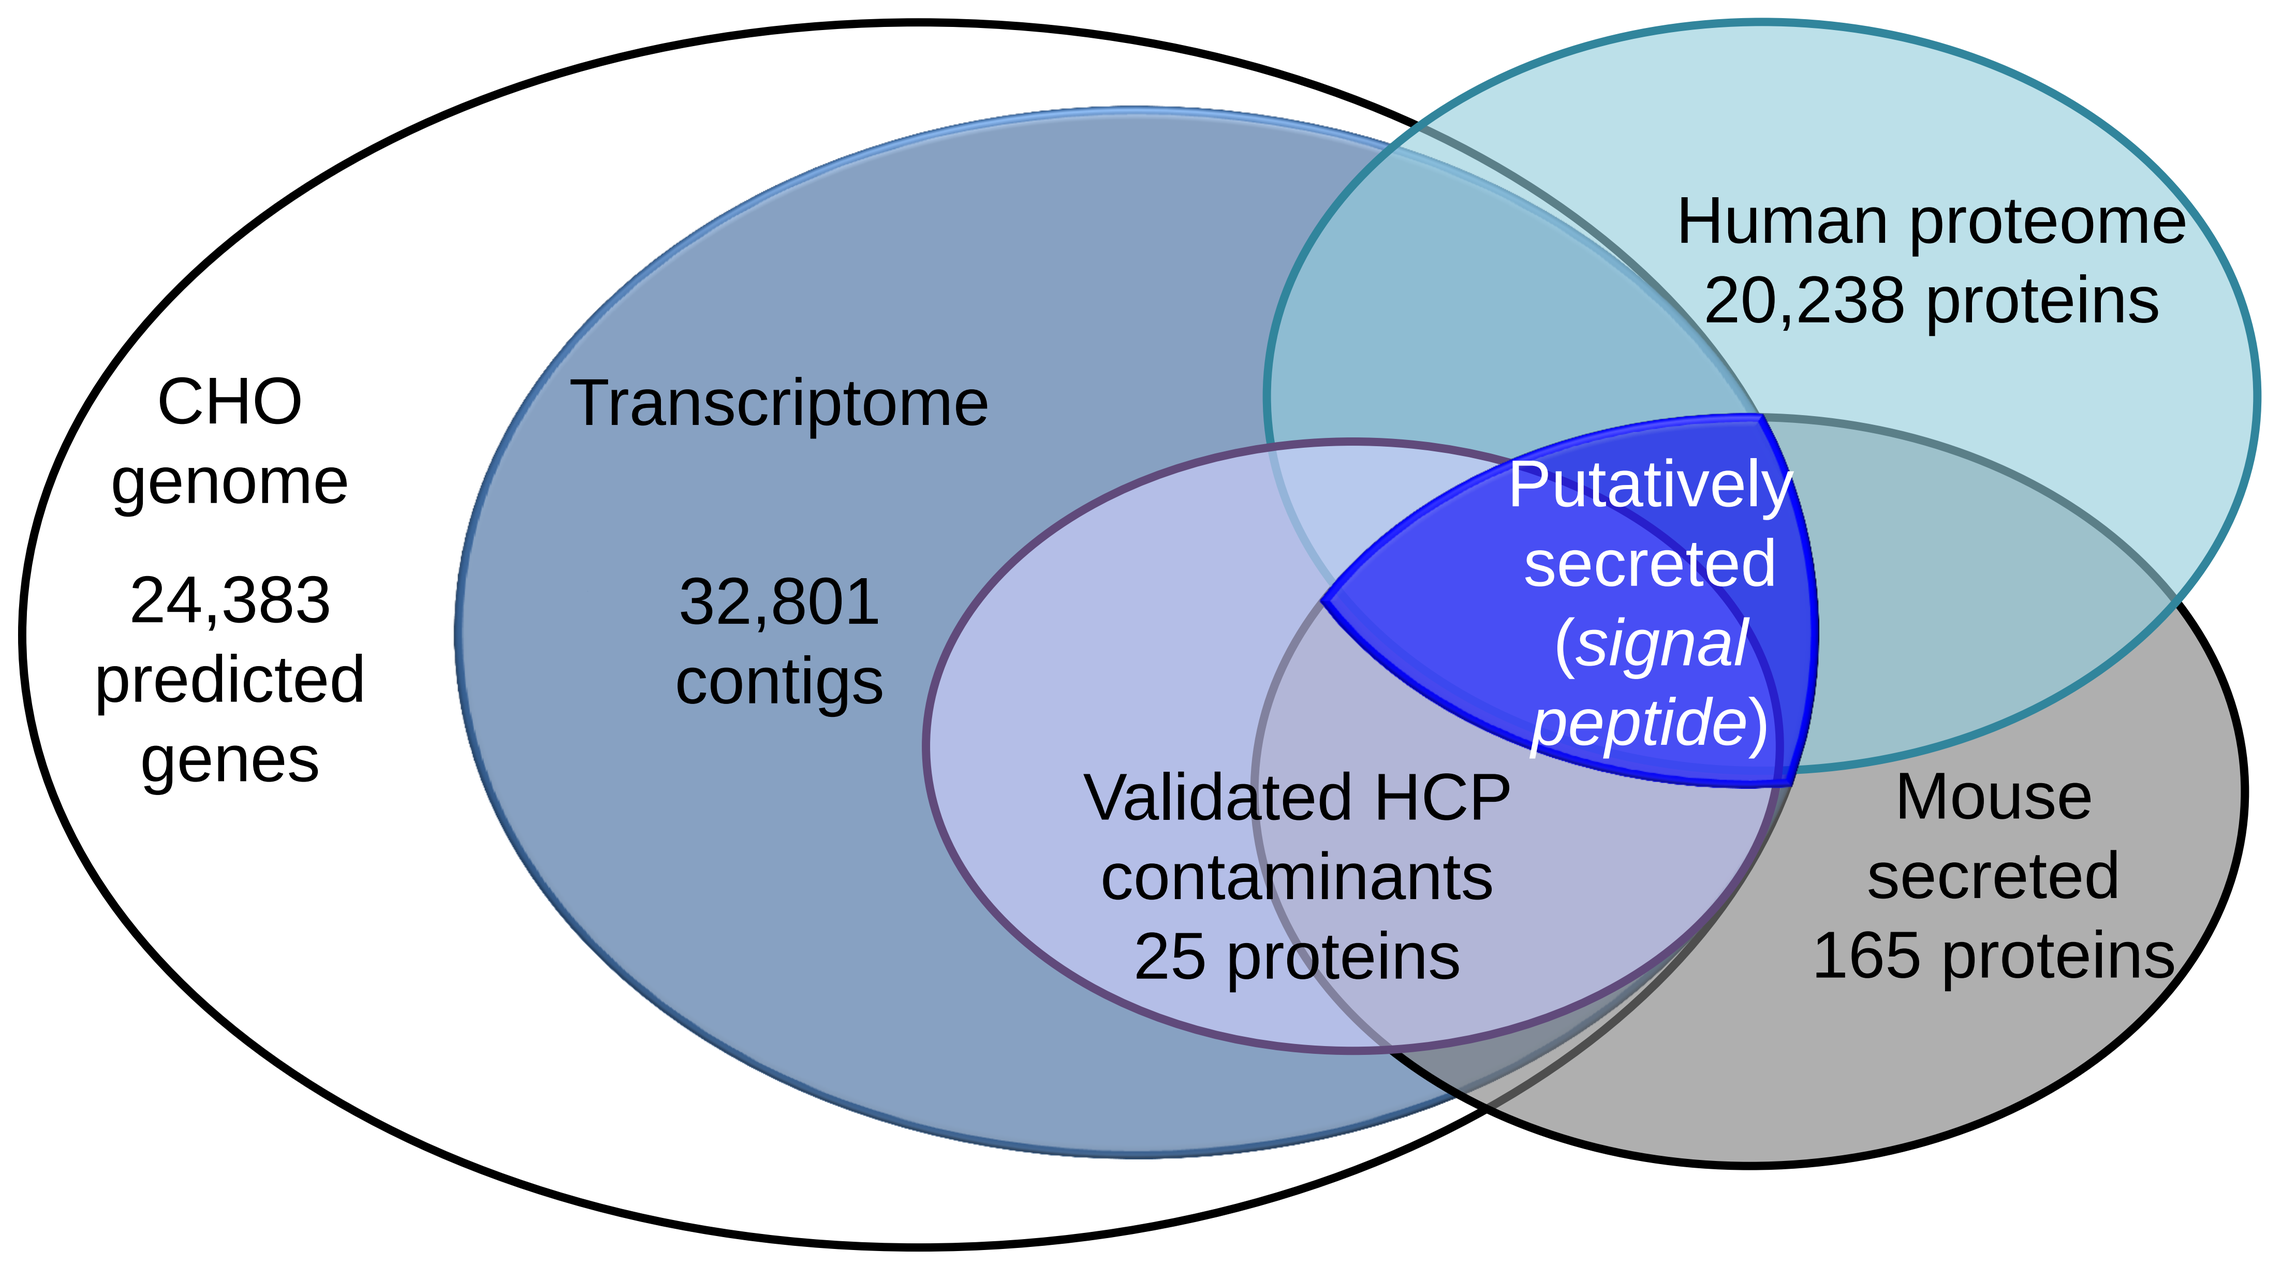

CHO genome
24,383 predicted genes
Transcriptome
32,801 contigs
Human proteome
20,238 proteins
Putatively secreted (signal peptide)
Validated HCP contaminants
25 proteins
Mouse secreted
165 proteins

Figure 2. Databases used for immunogenicity and cross-reactivity analysis. *Illustration of the sources used to analyzed potential CHO HCP immunogenicity Sources include CHO genome, CHO transcriptome, published CHO proteins identified in biotherapeutic products (Experimental HCP contaminants), and mouse secreted proteins downloaded from LOCATE and UniProtKB/Swiss-Prot.*

Immunogenicity Analysis

Immunogenic potential of validated HCP contaminants was assessed by epitope density and ranked on an immunogenicity scale, which was developed to evaluate therapeutic proteins of interest against known immunogenic and non-immunogenic proteins.

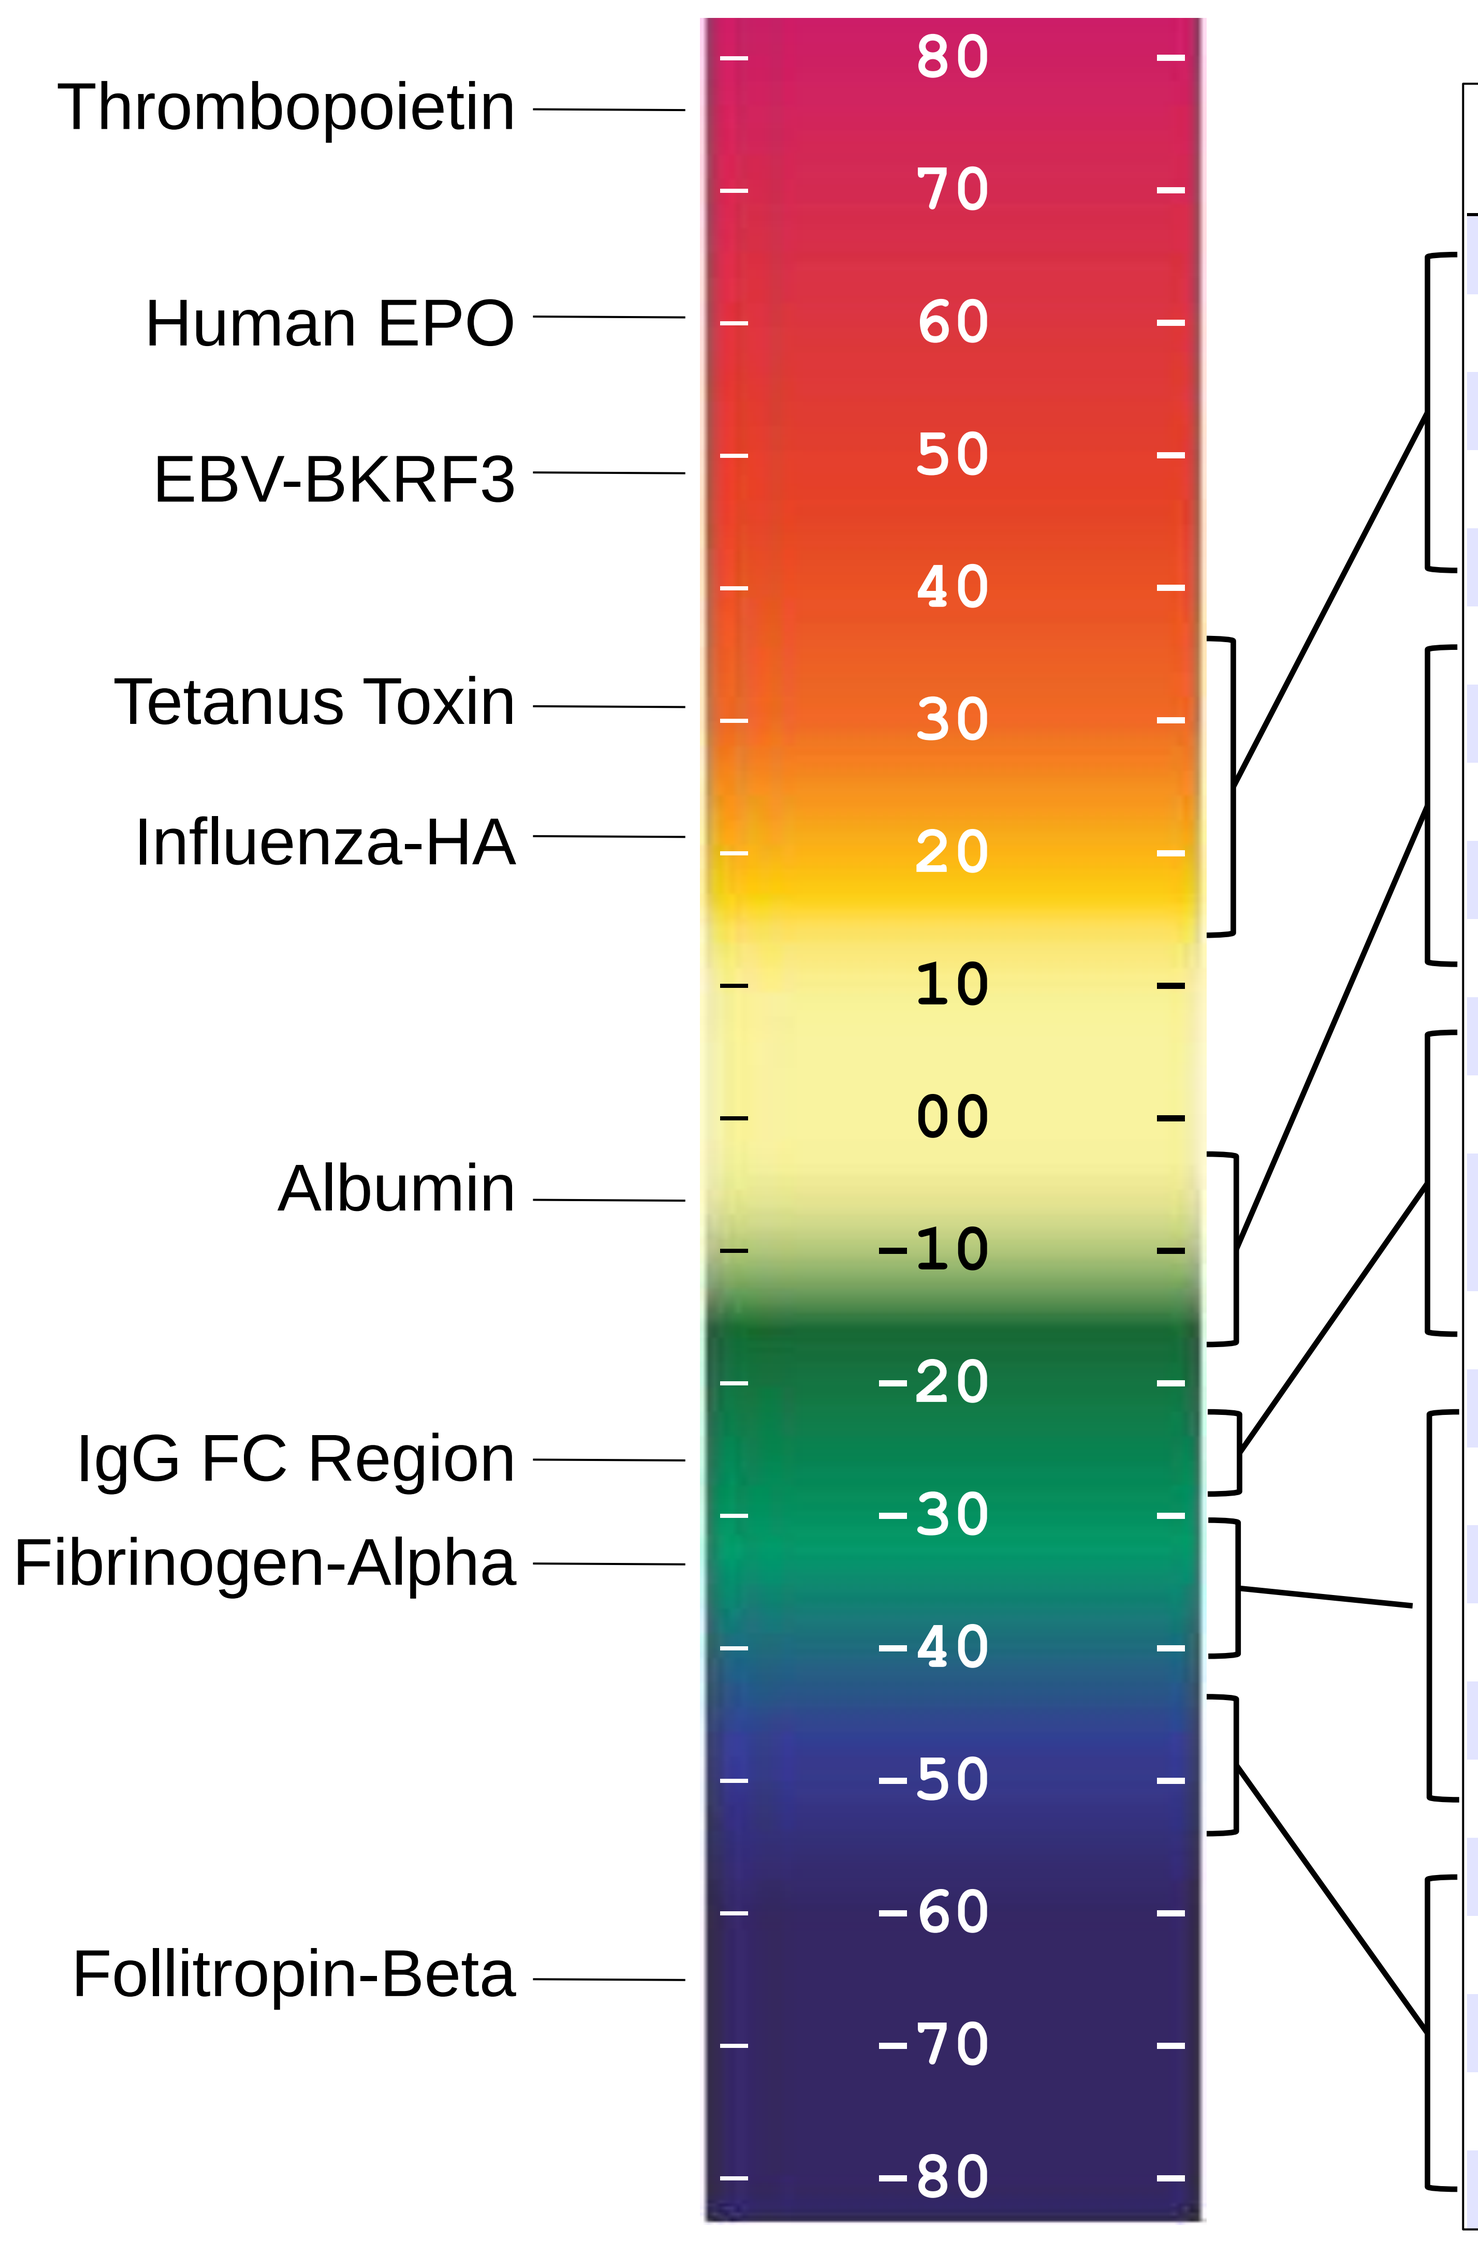

Thrombopoietin
Human EPO
EBV-BKRF3
Tetanus Toxin
Influenza-HA
Albumin
IgG FC Region
Fibrinogen-Alpha
Follitropin-Beta
80
70
60
50
40
30
20
10
00
-10
-20
-30
-40
-50
-60
-70
-80

protein	immuno.
Collagen alpha-1(III) chain	35.79
Lysosomal protective protein	34.4
Glutathione S-transferase P	25.16
Annexin A1	24.87
Annexin A2	14.87
Nuclear prelamin A recognition factor	-3.77
Actin, cytoplasmic 2	-9.36
Serine protease HTRA1	-13.02
Peptidyl-prolyl cis-trans isomerase B	-13.65
Macrophage-capping protein	-17.11
Vinculin	-22.45
Metalloproteinase inhibitor 2	-25.28
Procollagen C-endopeptidase enhancer 1	-28.49
Stathmin-4	-28.74
Elongation factor 2	-30.01
Transketolase	-33.53
Actin, cytoplasmic 1	-34.19
Dystroglycan	-36.63
Cathepsin Z	-39.59
Heat shock cognate 71 kDa protein	-39.63
Beta-2-microglobulin	-43.93
Galectin-1	-45.49
78 kDa glucose-regulated protein	-48.51
Elongation factor 2	-50.97
Peroxiredoxin-1	-51.76

Figure 3. EpiMatrix Protein Immunogenicity Scale. *Validated CHO HCP contaminants with scores ≥20 represent approximately the top 20% of the distribution. High-scoring proteins are considered potentially immunogenic. Conversely, proteins scoring below -20 are categorized as possessing "low" immunogenic potential. All scores normalized for protein length*

Immunogenicity comparison

- Immunogenicity scores calculated for proteins sourced from three different datasets.
- High-scoring proteins (above 20) are considered potentially immunogenic.
- Proteins scoring between -20 and 20 are less likely to generate a significant immune reaction.
- Major validated HCP contaminants peak co-locates with secreted proteins peak, suggesting lower potential immunogenicity for this set,
- A minor peak higher on the scale suggests that some HCP do carry a risk of immunogenicity.

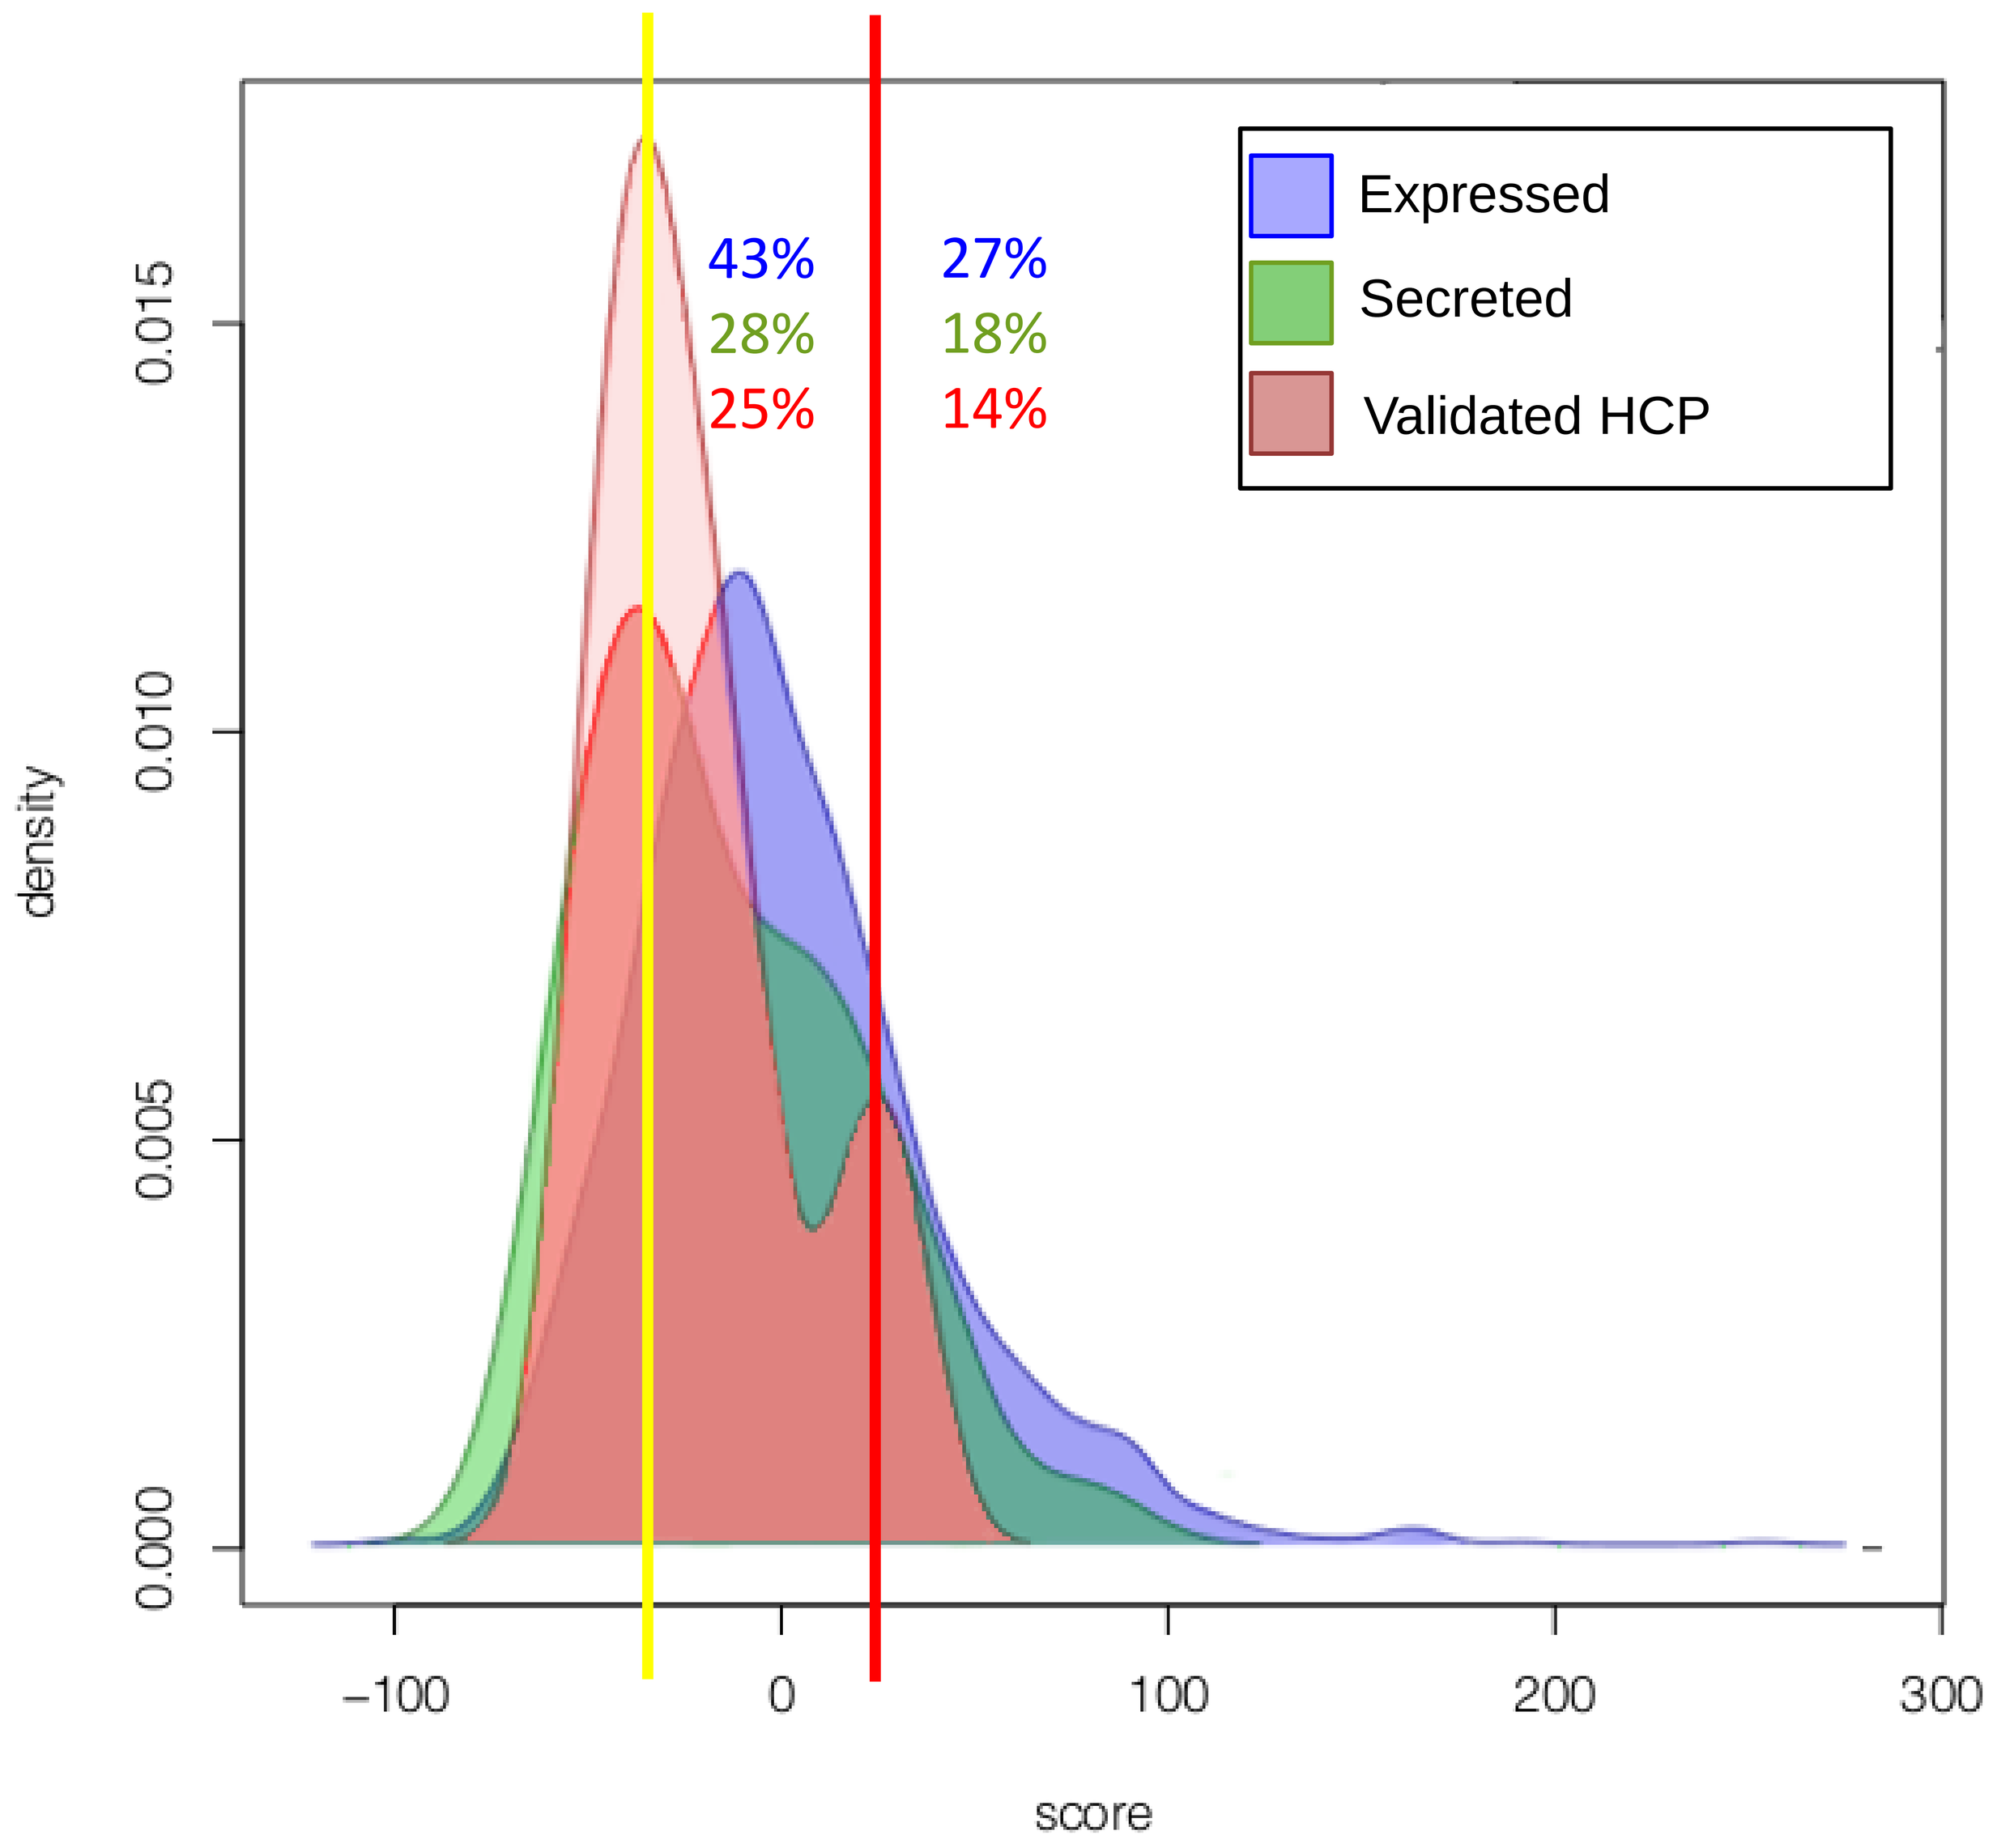

Expressed
Secreted
Validated HCP
43%
28%
25%
27%
18%
14%
density
0.000
0.005
0.010
0.015
-100
0
100
200
300
score

Figure 4. Kernel density estimate of potential and validated CHO HCP immunogenicity scores. *Shoulders in validated HCP contaminants (red) and putative secreted protein (green) sets arise due to higher subpopulation scores. Area under each curve is 1. Lines delimitate immunogenicity potential (yellow: neutral; red: high). Percentages are % predicted proteins with reduced (yellow to red lines) or increased (red line rightward) immunogenicity potential.*

CHOPPI Web Tool

CHOPPI (CHO Protein Predicted Immunogenicity) is a web tool developed to address two questions:

(1) **What are the possible contaminants?** CHOPPI integrates the genome of the CHO-K1 cell line, a CHO transcriptome, a mouse secretome, and SignalP-based signal peptide prediction (indicating likely secretion).

(2) **Which are likely to be immunogenic?** CHOPPI assesses predicted T cell epitope content by EpiMatrix, as well as compares the proteins to their human homologs and their predicted epitopes to predicted epitopes within the human genome.

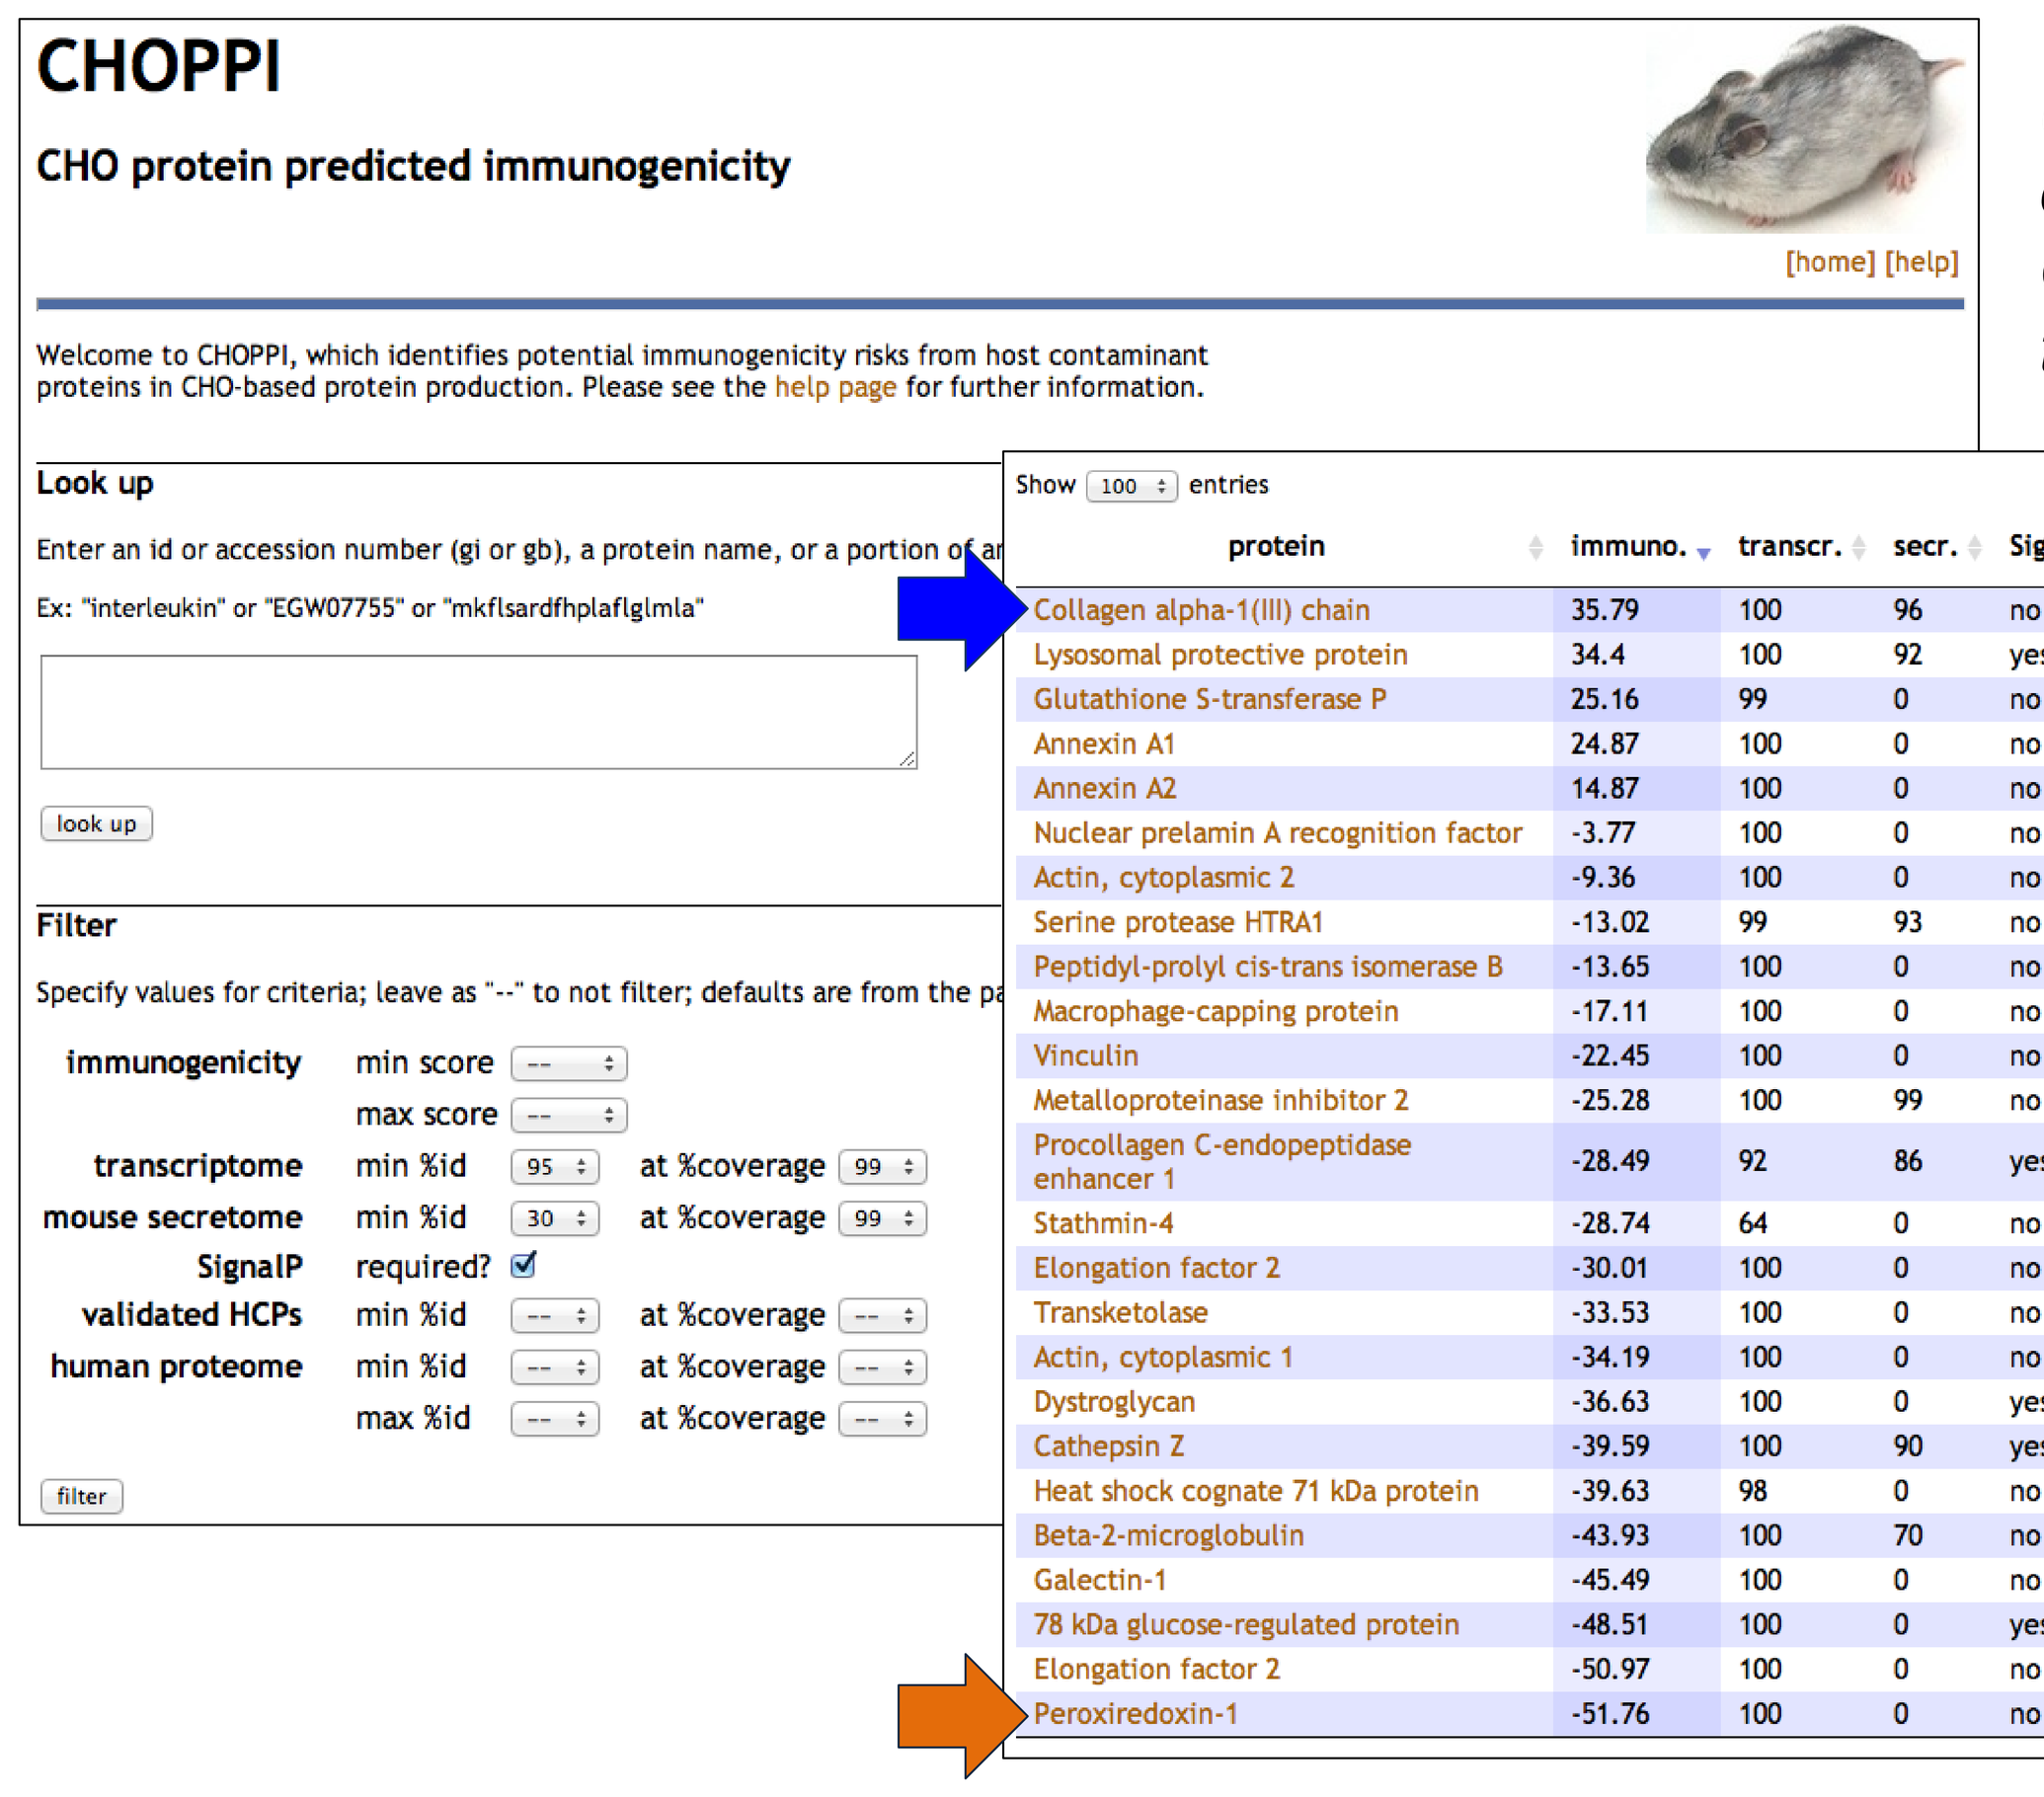

CHOPPI

CHO protein predicted immunogenicity

[home] [help]

Welcome to CHOPPI, which identifies potential immunogenicity risks from host contaminant proteins in CHO-based protein production. Please see the help page for further information.

Look up

Enter an id or accession number (gi or gb), a protein name, or a portion of an

Ex: "interleukin" or "EGW07755" or "mkflsardfhplaflglmla"

look up

Filter

Specify values for criteria; leave as "--" to not filter; defaults are from the pa

immunogenicity min score -- max score --

transcriptome min %id 95 at %coverage 99

mouse secretome min %id 30 at %coverage 99

SignalP required?

validated HCPs min %id -- at %coverage --

human proteome min %id -- at %coverage -- max %id -- at %coverage --

filter

Show 100 entries Search:

protein	immuno.	transcr.	secr.	SignalP	validated	human	9mers	unique 9mers
Collagen alpha-1(III) chain	35.79	100	96	no	100	92	228	60
Lysosomal protective protein	34.4	100	92	yes	100	87	109	28
Glutathione S-transferase P	25.16	99	0	no	100	87	45	14
Annexin A1	24.87	100	0	no	100	89	75	20
Annexin A2	14.87	100	0	no	100	97	39	4
Nuclear prelamin A recognition factor	-3.77	100	0	no	100	83	54	13
Actin, cytoplasmic 2	-9.36	100	0	no	100	100	39	0
Serine protease HTRA1	-13.02	99	93	no	100	91	38	6
Peptidyl-prolyl cis-trans isomerase B	-13.65	100	0	no	100	94	31	5
Macrophage-capping protein	-17.11	100	0	no	100	93	50	5
Vinculin	-22.45	100	0	no	100	98	164	3
Metalloproteinase inhibitor 2	-25.28	100	99	no	100	93	29	1
Procollagen C-endopeptidase enhancer 1	-28.49	92	86	yes	100	72	66	23
Stathmin-4	-28.74	64	0	no	100	99	27	1
Elongation factor 2	-30.01	100	0	no	100	99	131	3
Transketolase	-33.53	100	0	no	100	95	87	11
Actin, cytoplasmic 1	-34.19	100	0	no	100	100	49	0
Dystroglycan	-36.63	100	0	yes	100	93	121	18
Cathepsin Z	-39.59	100	90	yes	100	84	36	11
Heat shock cognate 71 kDa protein	-39.63	98	0	no	100	98	29	5
Beta-2-microglobulin	-43.93	100	70	no	100	70	4	3
Galectin-1	-45.49	100	0	no	100	91	16	2
78 kDa glucose-regulated protein	-48.51	100	0	yes	100	99	86	4
Elongation factor 2	-50.97	100	0	no	100	100	8	0
Peroxiredoxin-1	-51.76	100	0	no	100	95	24	5

Users may retrieve lists of proteins from the CHO genome according to specified filters.

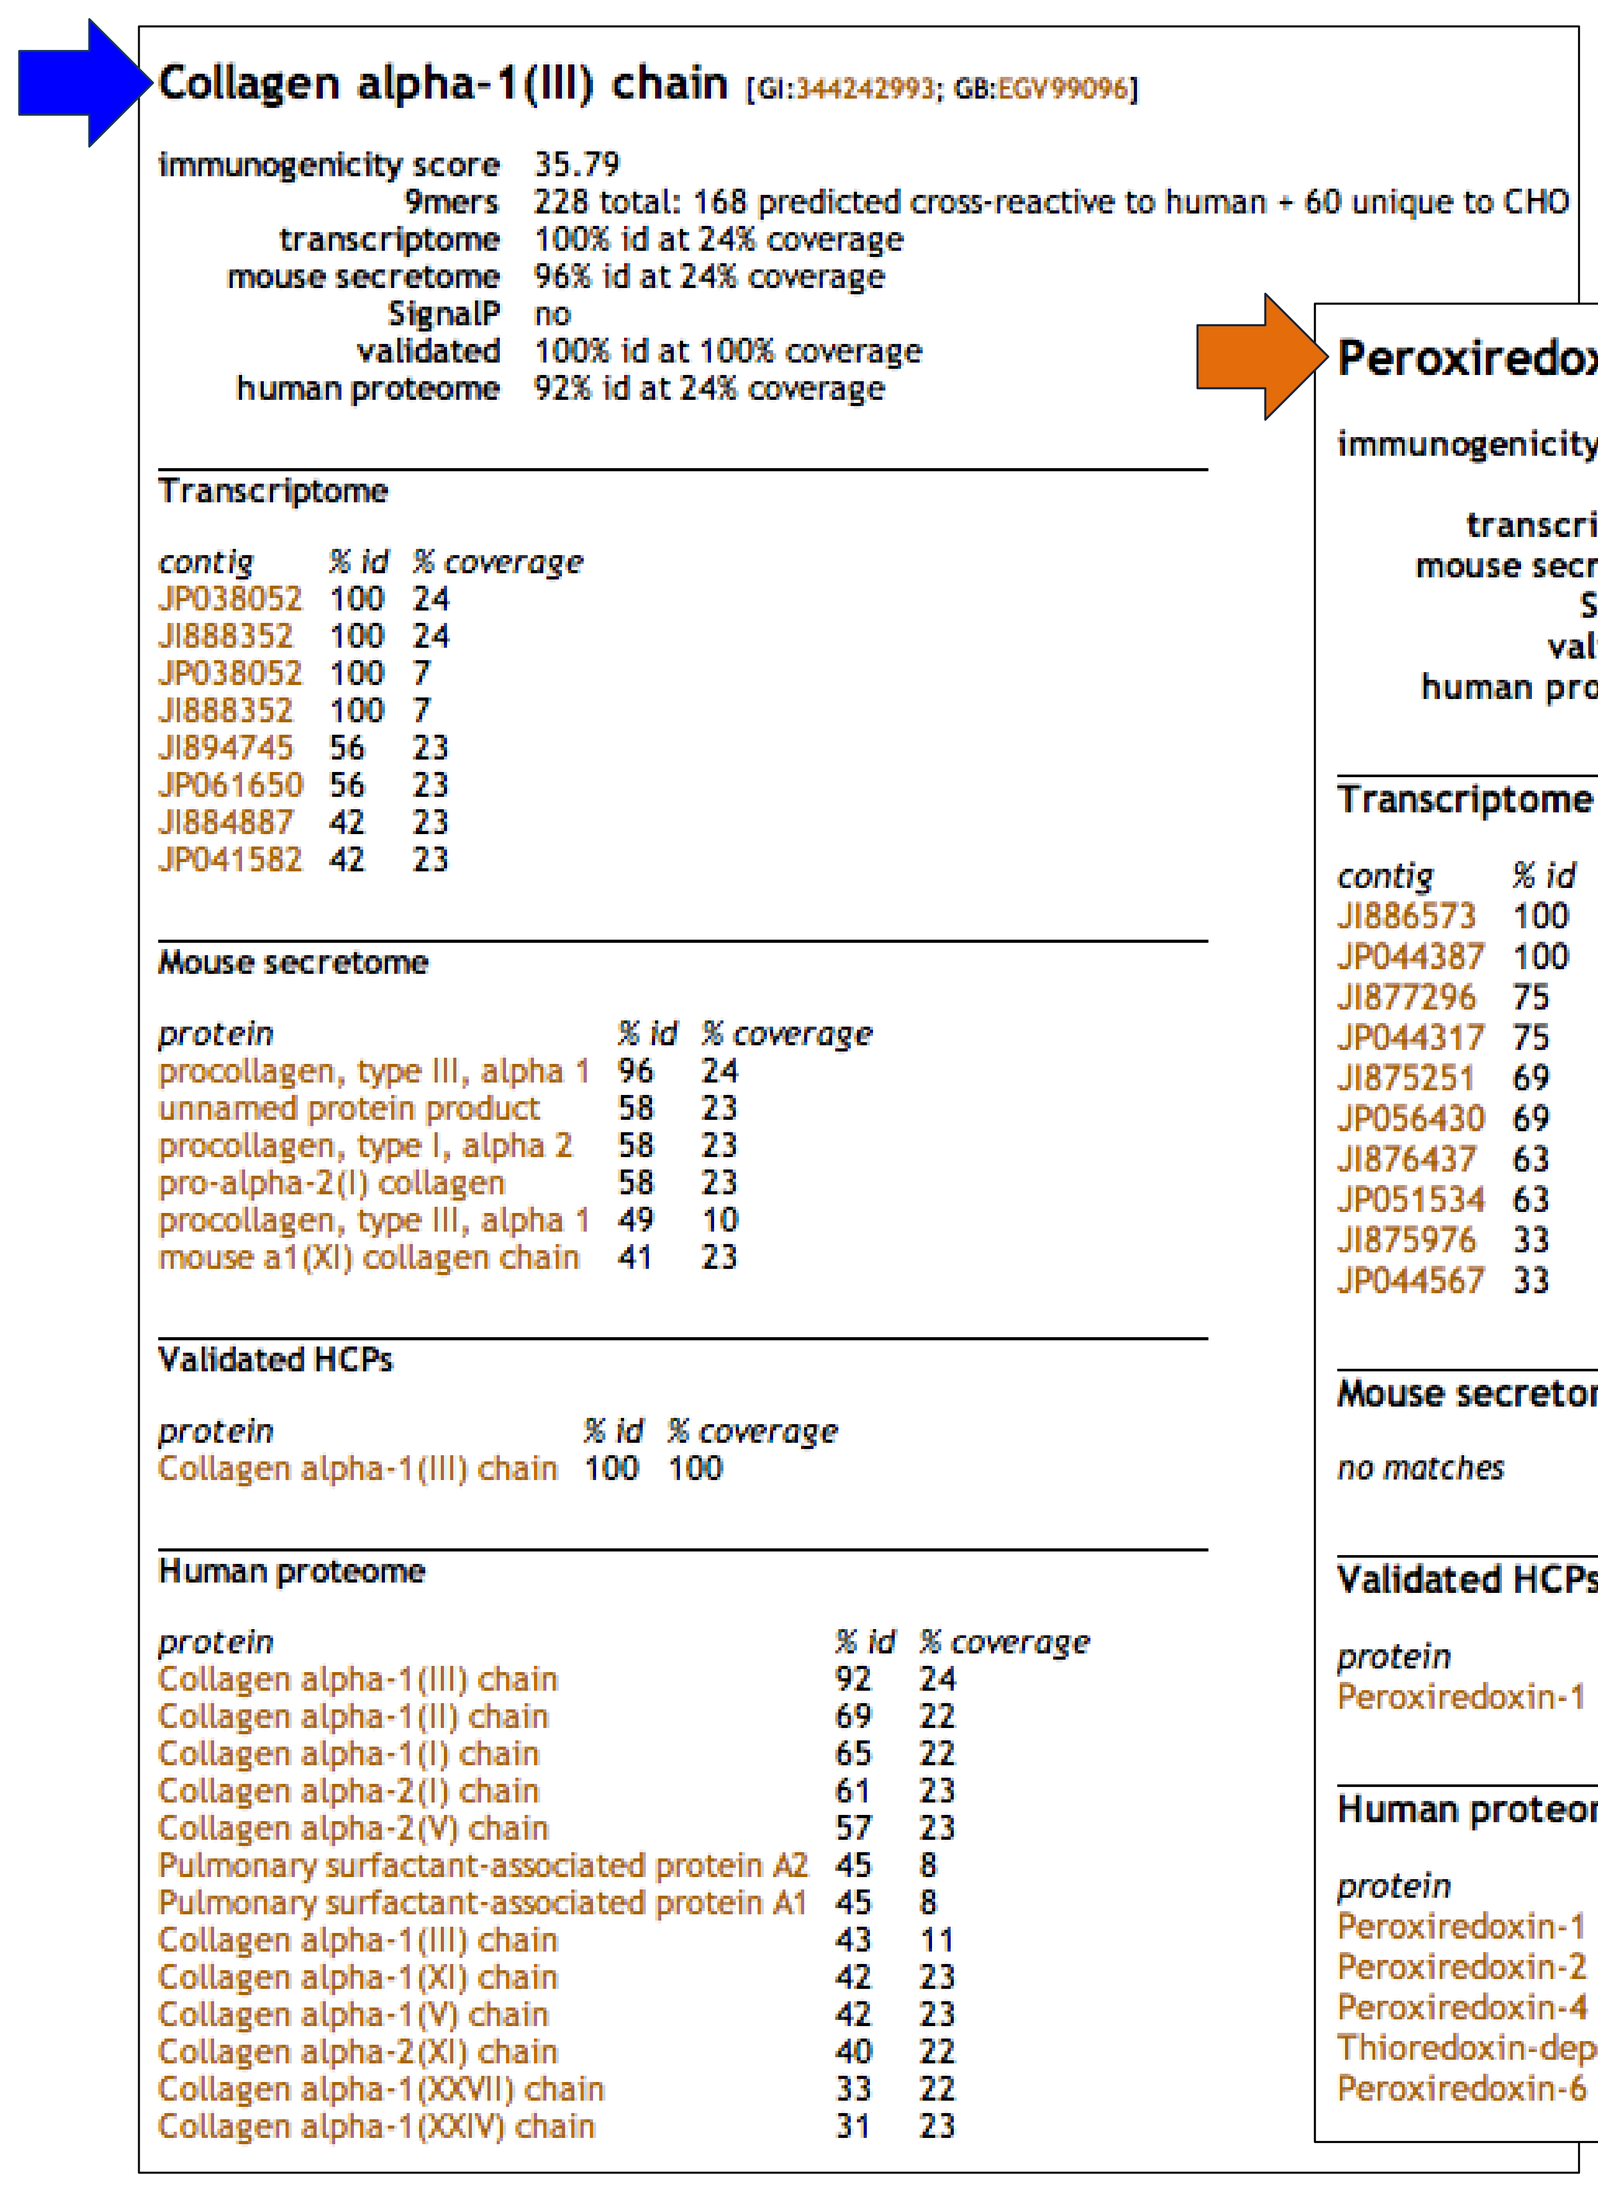

Collagen alpha-1(III) chain [GI:344242993; GB:EGV99096]

immunogenicity score 35.79
9mers 228 total: 168 predicted cross-reactive to human + 60 unique to CHO
transcriptome 100% id at 24% coverage
mouse secretome 96% id at 24% coverage
SignalP no
validated 100% id at 100% coverage
human proteome 92% id at 24% coverage

Transcriptome

contig	% id	% coverage
JP038052	100	24
JI888352	100	24
JP038052	100	7
JI888352	100	7
JI894745	56	23
JP061650	56	23
JI884887	42	23
JP041582	42	23

Mouse secretome

protein	% id	% coverage
procollagen, type III, alpha 1	96	24
unnamed protein product	58	23
procollagen, type I, alpha 2	58	23
pro-alpha-2(I) collagen	58	23
procollagen, type III, alpha 1	49	10
mouse a1(XI) collagen chain	41	23

Validated HCPs

protein	% id	% coverage
Collagen alpha-1(III) chain	100	100

Human proteome

protein	% id	% coverage
Collagen alpha-1(III) chain	92	24
Collagen alpha-1(II) chain	69	22
Collagen alpha-1(I) chain	65	22
Collagen alpha-2(I) chain	61	23
Collagen alpha-2(V) chain	57	23
Pulmonary surfactant-associated protein A2	45	8
Pulmonary surfactant-associated protein A1	45	8
Collagen alpha-1(III) chain	43	11
Collagen alpha-1(XI) chain	42	23
Collagen alpha-1(V) chain	42	23
Collagen alpha-2(XI) chain	40	22
Collagen alpha-1(XXVII) chain	33	22
Collagen alpha-1(XXIV) chain	31	23

The tool presents protein sets in HTML tables sortable by column and allows export to CSV-format files.

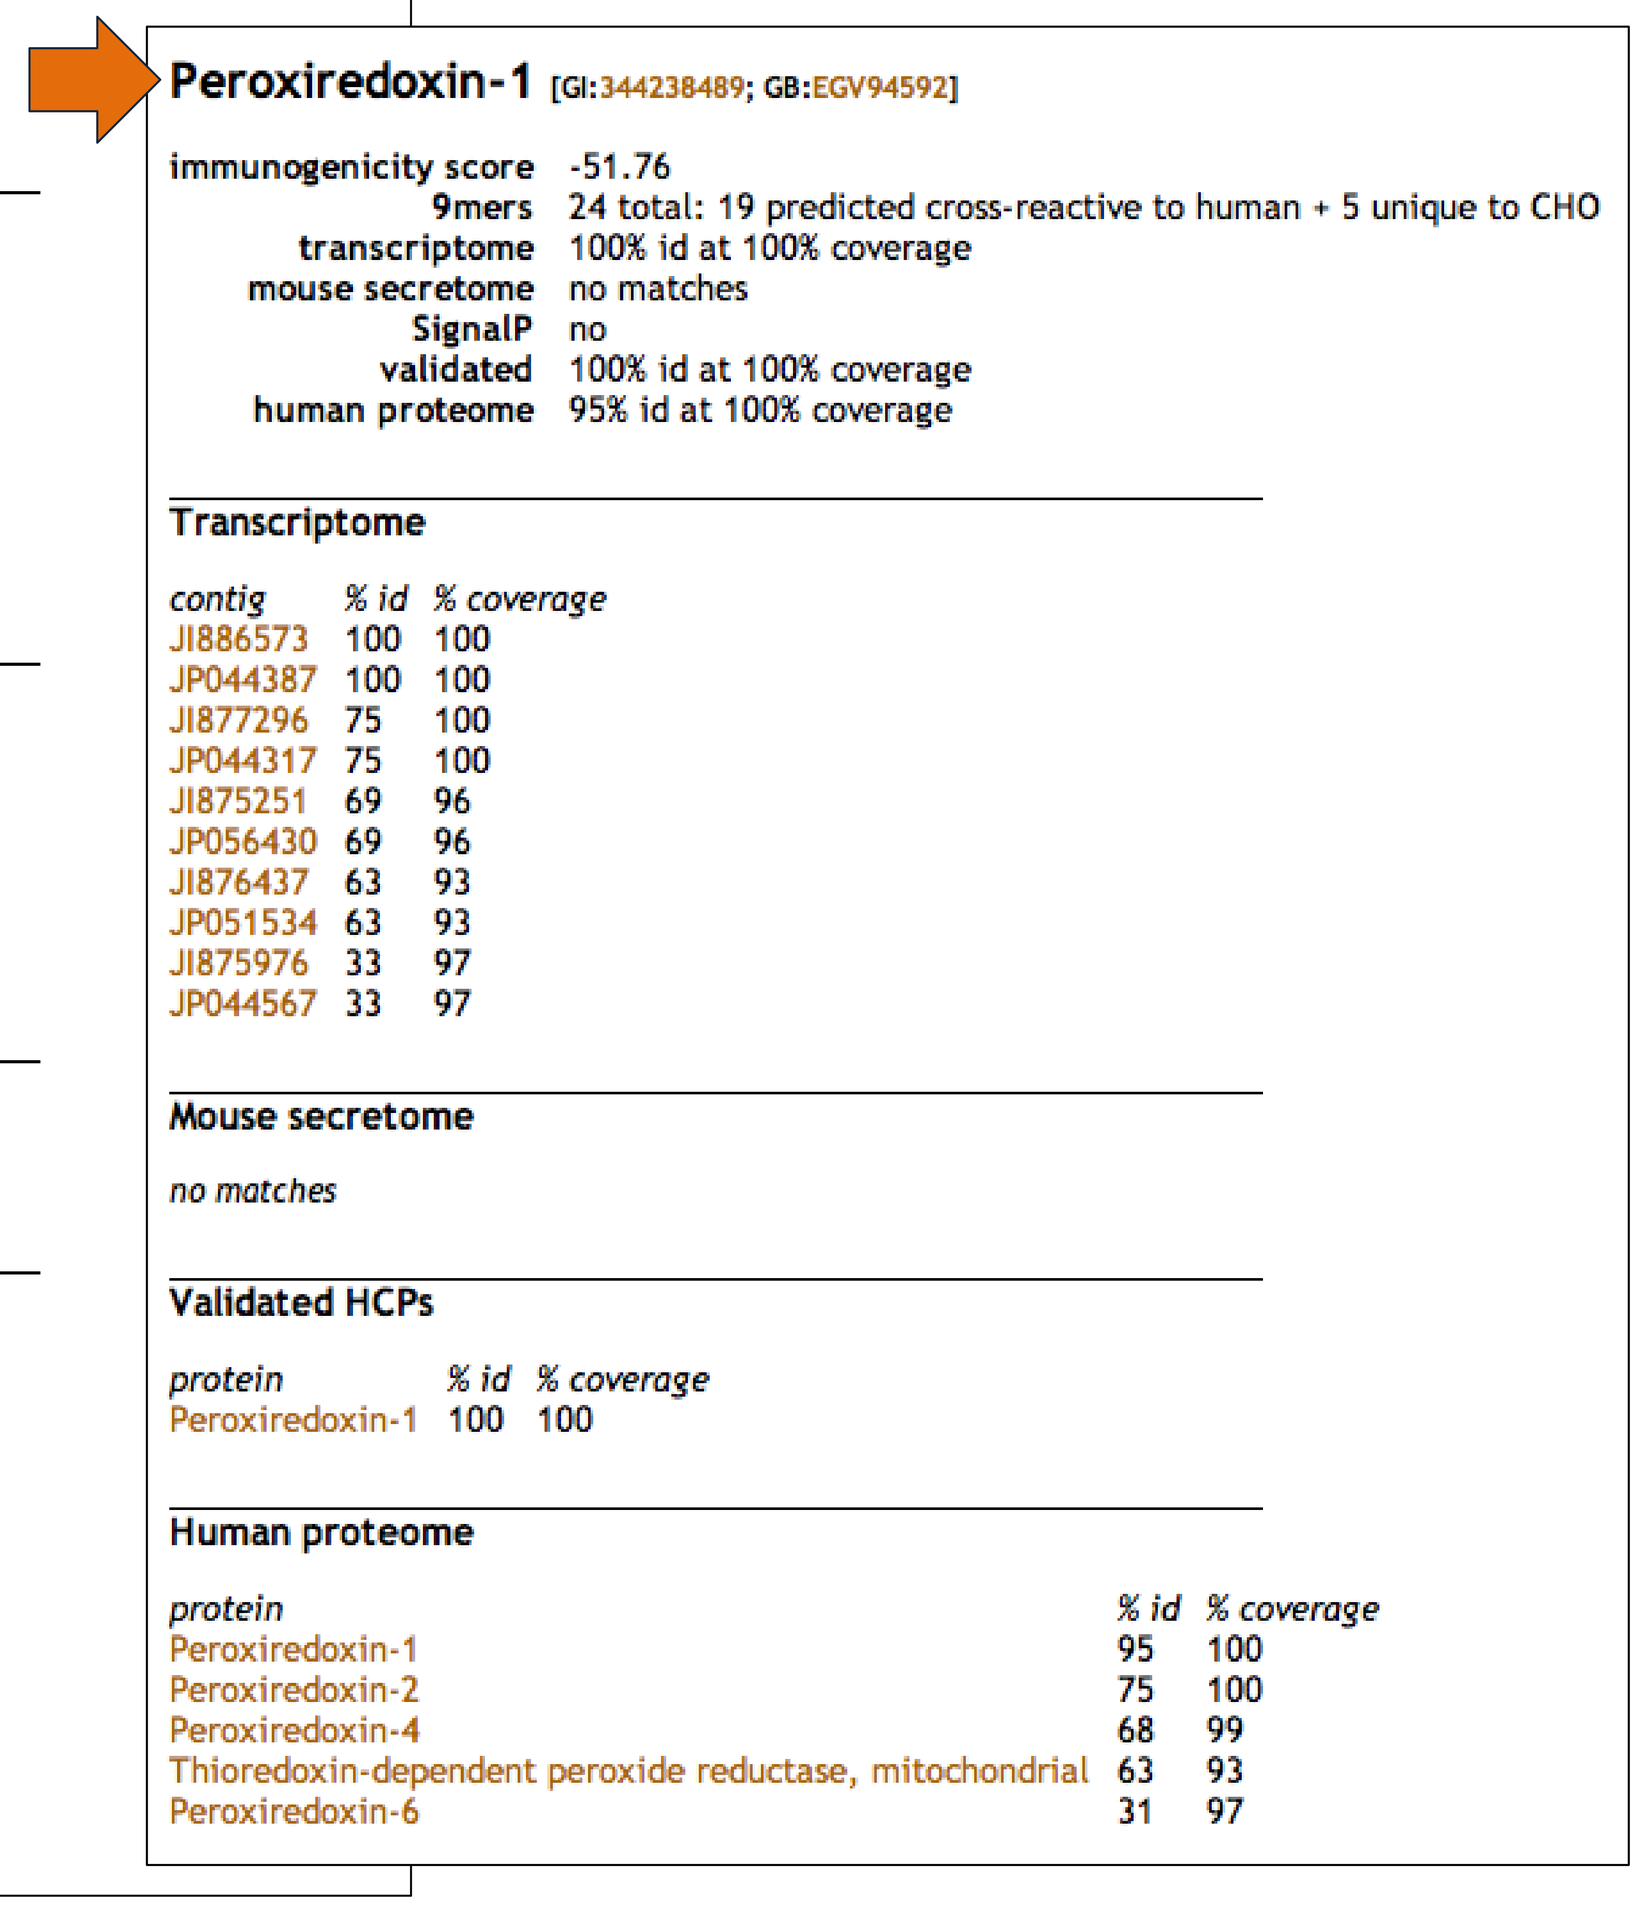

Peroxiredoxin-1 [GI:344238489; GB:EGV94592]

immunogenicity score -51.76
9mers 24 total: 19 predicted cross-reactive to human + 5 unique to CHO
transcriptome 100% id at 100% coverage
mouse secretome no matches
SignalP no
validated 100% id at 100% coverage
human proteome 95% id at 100% coverage

Transcriptome

contig	% id	% coverage
JI886573	100	100
JP044387	100	100
JI877296	75	100
JP044317	75	100
JI875251	69	96
JP056430	69	96
JI876437	63	93
JP051534	63	93
JI875976	33	97
JP044567	33	97

Mouse secretome

no matches

Validated HCPs

protein	% id	% coverage
Peroxiredoxin-1	100	100

Human proteome

protein	% id	% coverage
Peroxiredoxin-1	95	100
Peroxiredoxin-2	75	100
Peroxiredoxin-4	68	99
Thioredoxin-dependent peroxide reductase, mitochondrial	63	93
Peroxiredoxin-6	31	97

Figure 5. CHOPPI web tool (http://clover.cs.dartmouth.edu/~cbk/choppi/). *Each protein is characterized in terms of homology criteria and signal peptide prediction, along with assessments of epitope count and density, and count and density of epitopes unique to CHO according to a human cross-reactivity analysis.*

Conclusions

- The presence of HCP in therapeutic protein products may contribute to adverse clinical side effects and may impact drug efficacy.
- The impact of cross-conserved epitopes on human immune responses may include induction of effector T cells, induction of Tregs, or anergy.
- Our ability to score HCP for immunogenicity opens the door for future analysis such as the screening of whole proteins from the CHO transcriptome.
- Future HCP immunogenicity scores will be adjusted based on the prevalence of the human homolog in human extracellular fluids (such as serum).
- In vitro studies will be required to determine the impact of our findings and their implications.

Discussions leading to the development of CHOPPI were supported by NIH U19 grant AI082642 (to ADG). We thank our collaborators at iCubed, EpiVax, and Dartmouth for their contributions to this work.