Protective swine influenza T cell epitopes can be identified using PigMatrix and are conserved in field strains from 2013 to 2017

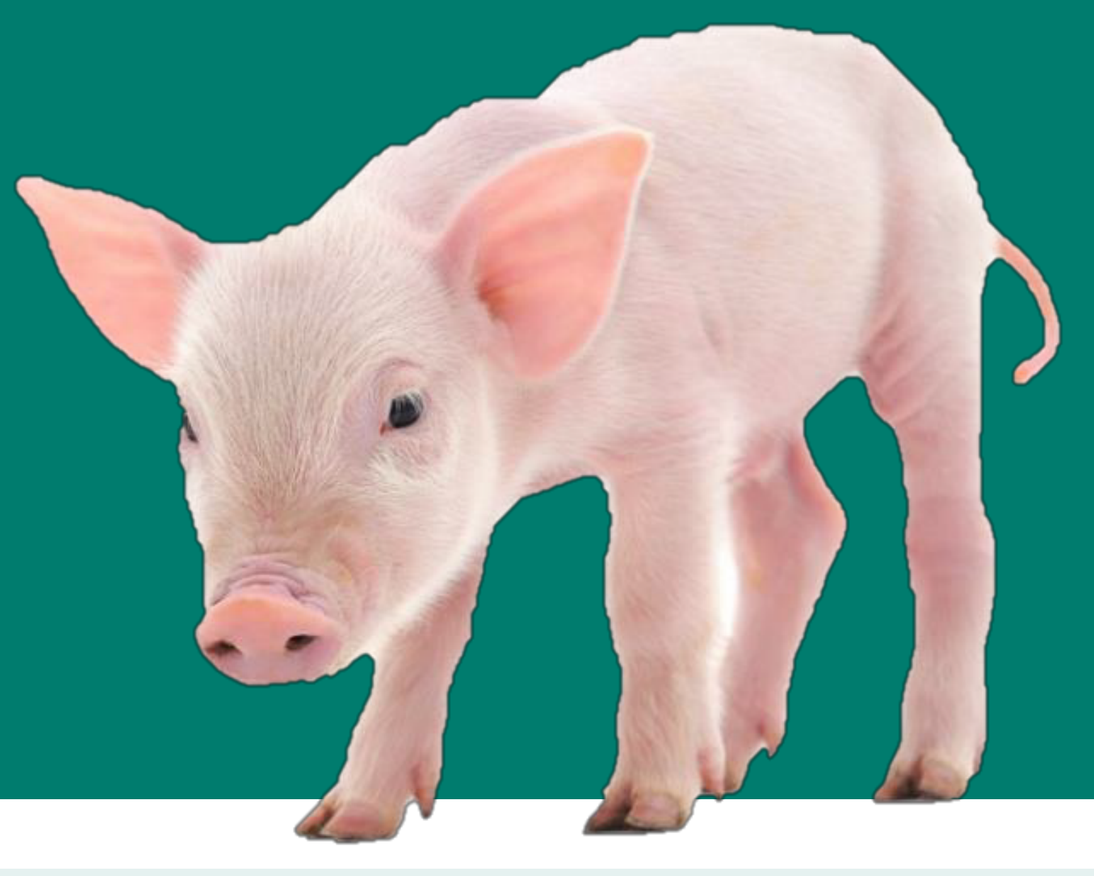

Quantifying the persistence of vaccine T cell epitopes in circulating swine influenza strains from 2013 - 2017

S. Tan[1], A. H. Gutierrez[2], P. C. Gauger[3], T. Opriessnig[3], L. Moise[1,2], A. S. De Groot[1,2]

[1]Institute for Immunology and Informatics, University of Rhode Island, Kingston, RI, [2]EpiVax, Inc., Providence, RI,
[3]Veterinary Diagnostic and Production Animal Medicine, College of Veterinary Medicine, Iowa State University, Ames, IA

Introduction

When swine influenza vaccine and circulating strains are poorly matched, vaccine-induced antibodies may not protect, but highly conserved T-cell epitopes may still have a disease-mitigating effect. This is T cell-mediated protection.

Challenge

Can I predict if my vaccine will induce T cell-mediated protection against a new flu strain?

Rationale

Higher conservation = higher protection.

Conservation of T cell epitopes between vaccine and novel swine influenza A virus (IAV) strains is variable and may explain variability in vaccine efficacy.

Approach

Analyze the degree of epitope cross-conservation between a **T cell-directed DNA vaccine** (class I and II) and **1272 swine influenza** outbreak strains **over five years** (Figure 1) by **immunoinformatics** methods (Figure 2).

Results

Cross-conserved T cell epitopes are crucial to confer protection as viral sequence similarity changes from year to year (Figure 3). Whole proteome EpiCC score of all IAV field strains suggests that the epitope-based DNA vaccine is able to drive better CD4+ immune response by having cross-conserved T cell epitope content (Figure 4). Consistency of the area under the curve (AUC) relative to 2013 implies the T cell epitope in the prototype vaccine should be able to drive good immune response over time (Figure 5).

Conclusions

The higher the EpiCC score, the higher T cell epitope content relatedness to the vaccine of interest. This case study shows good coverage of flu strains by a single vaccine over many years.

Figure 1| Availability of swine IAV sequences from 2013 to 2017

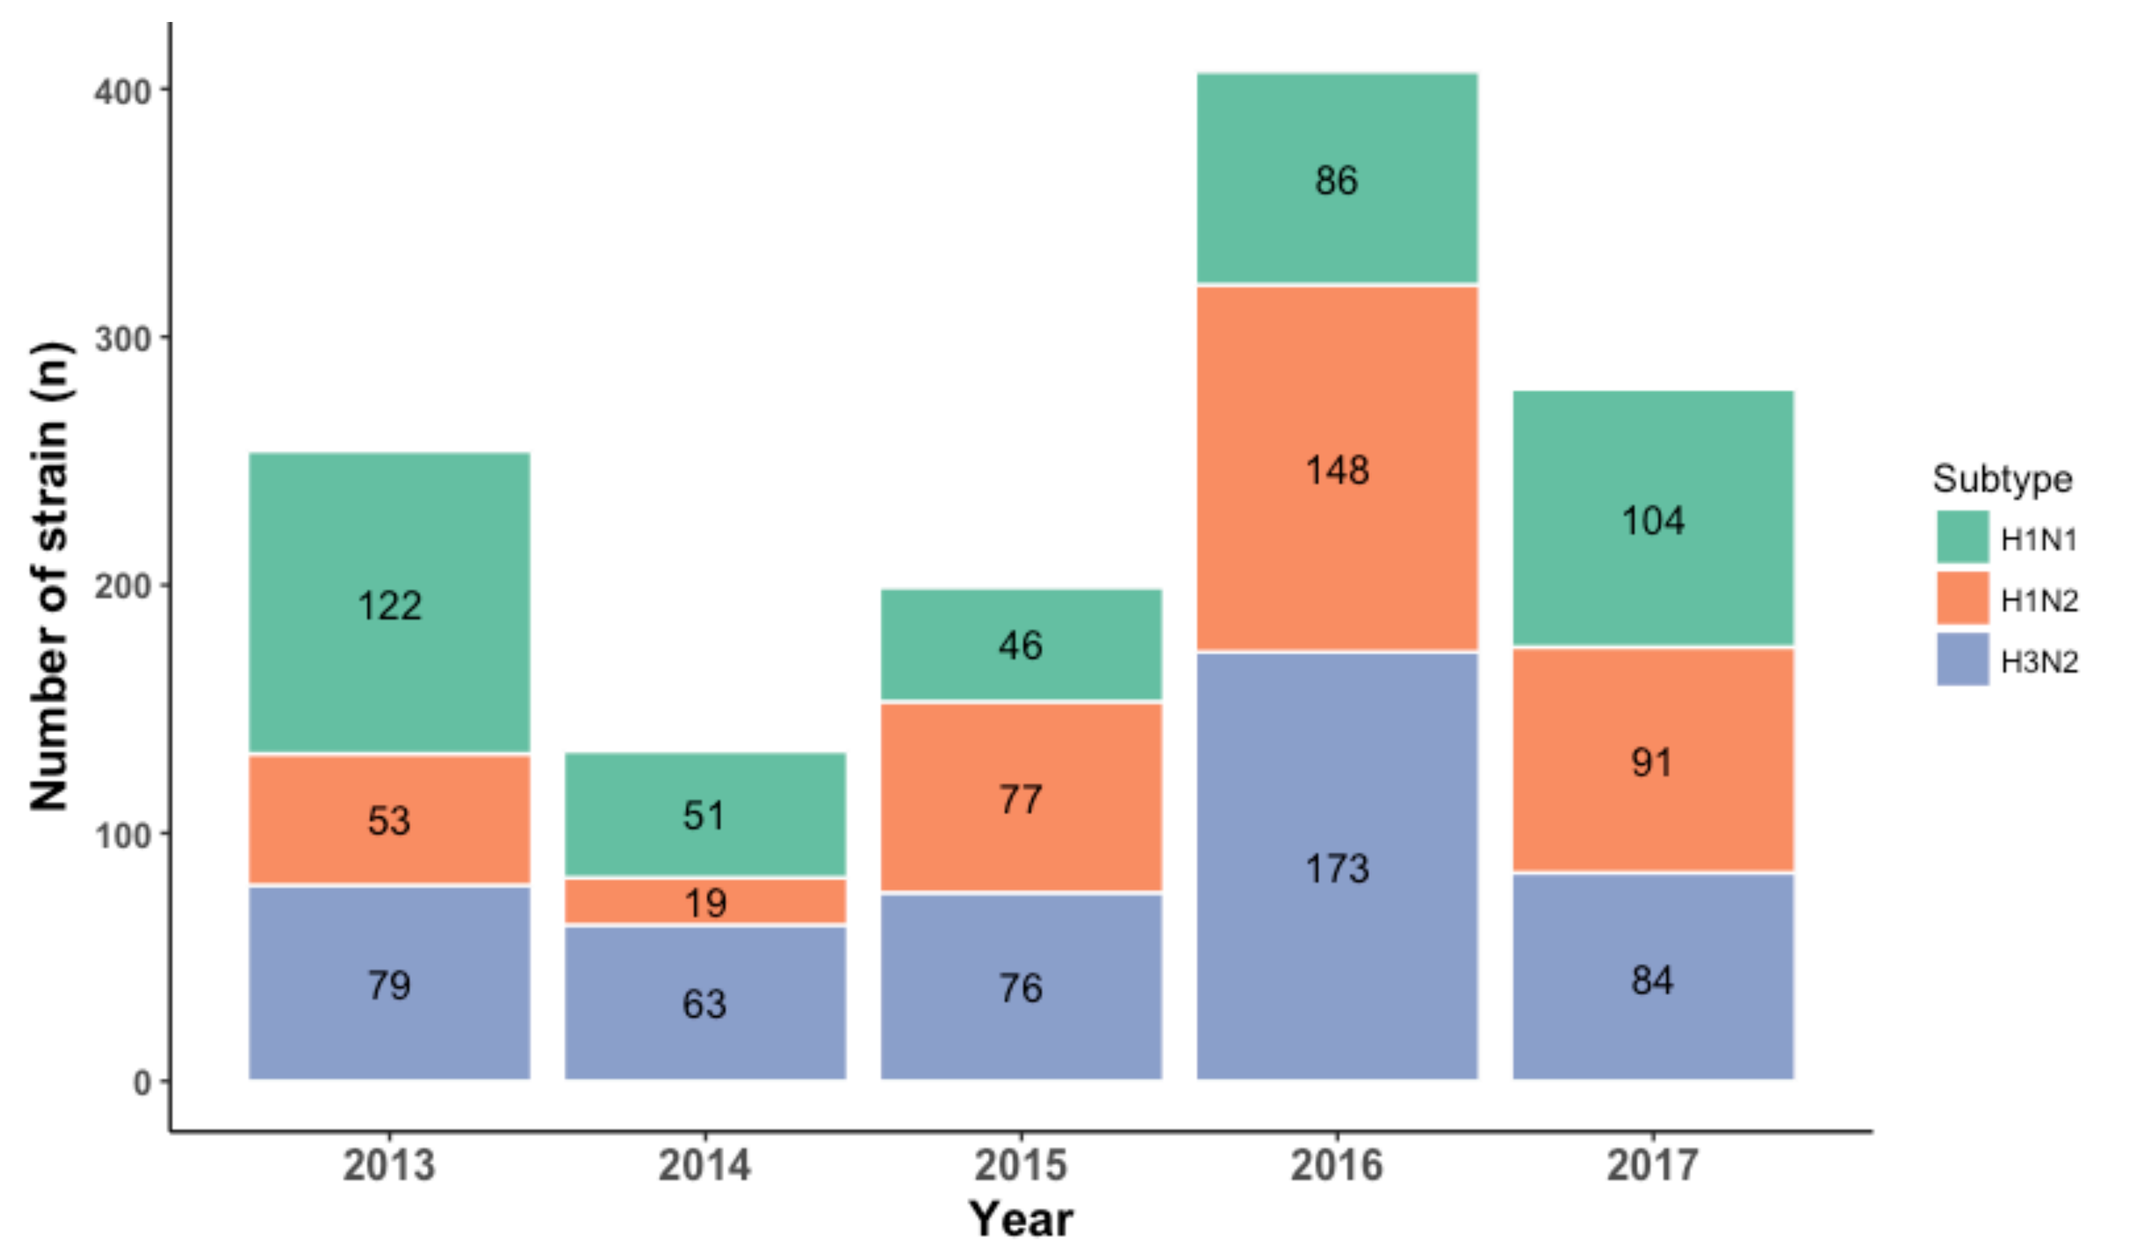
Number of strain (n)
400
300
200
100
0
122
53
79
51
19
63
46
77
76
86
148
173
104
91
84
2013
2014
2015
2016
2017
Year
Subtype
H1N1
H1N2
H3N2

Figure 2| Summary of analyses flow and immunoinformatics tools

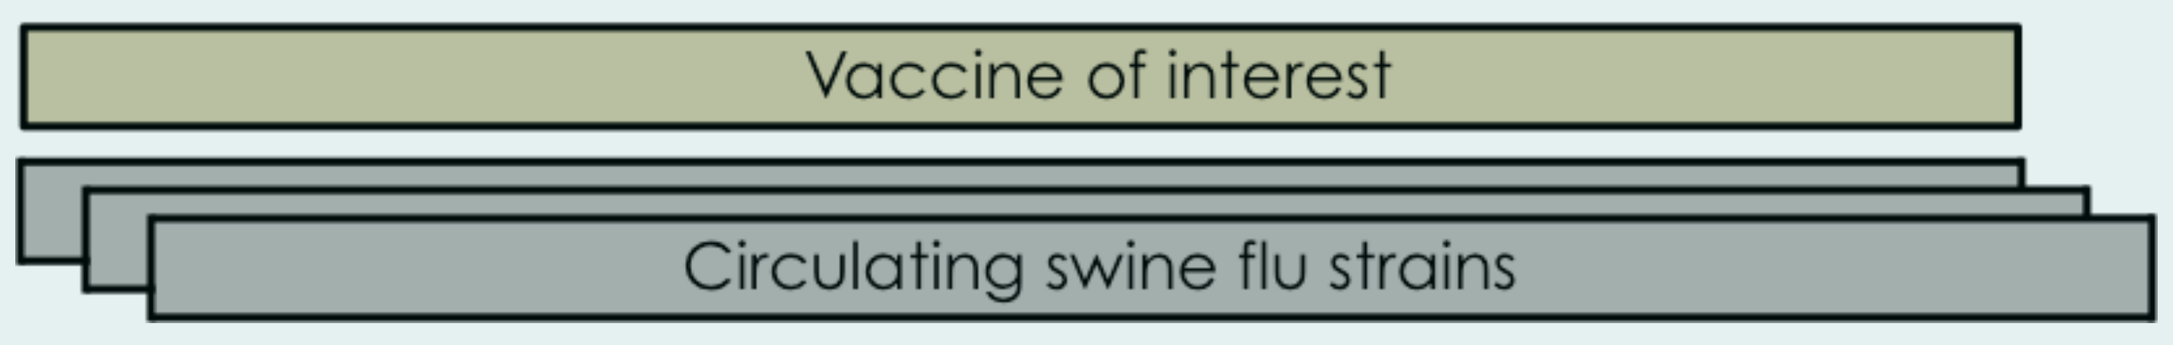
Vaccine of interest
Circulating swine flu strains

Conservation analysis (Conservatrix)

- 9-mer sequences were searched for identically matched segments and ranked by peptide conservation among IAV strains

T cell epitope prediction (PigMatrix)

- Matrices that are built based on the pocket profile method using EpiMatrix **pocket profiles** for HLA epitope prediction and were designed based on the binding preferences of the best-matched HLA pocket for each **SLA pocket**.
- PigMatrix raw scores were standardized to Z-scores to compare potential epitopes across multiple SLA alleles. Peptides with Z-scores ≥ 1.64 (the top 5% of any given sample of 9-mers) were identified as likely to be SLA ligands.

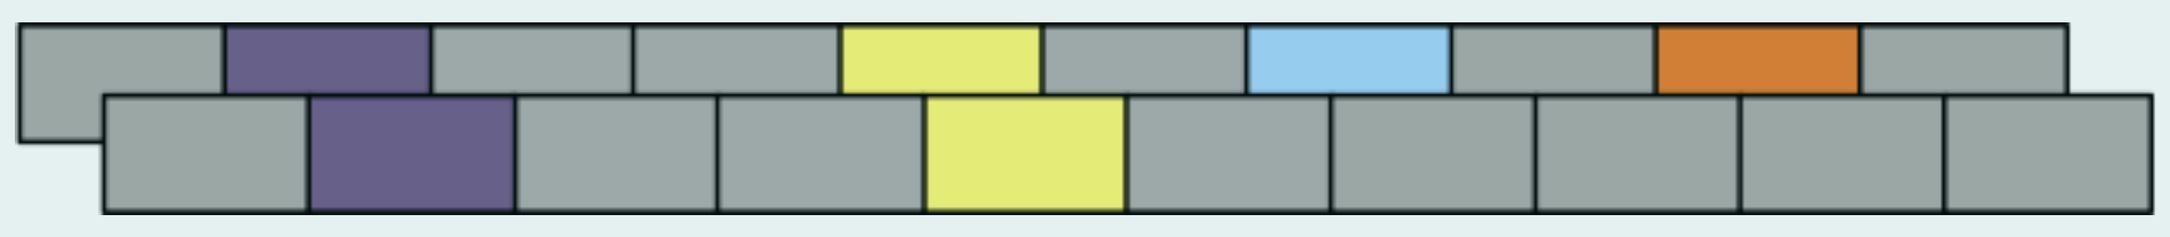

Conservation at the TCR face (JanusMatrix)

- At the level of an SLA-bound peptide, JanusMatrix is applied to identify PigMatrix-predicted epitopes that are in contact with the SLA molecule itself while others are accessible to the TCR.
- Each epitope has two faces: a TCR-binding face (**green** amino acids); a SLA-binding face (**blue** amino acids).

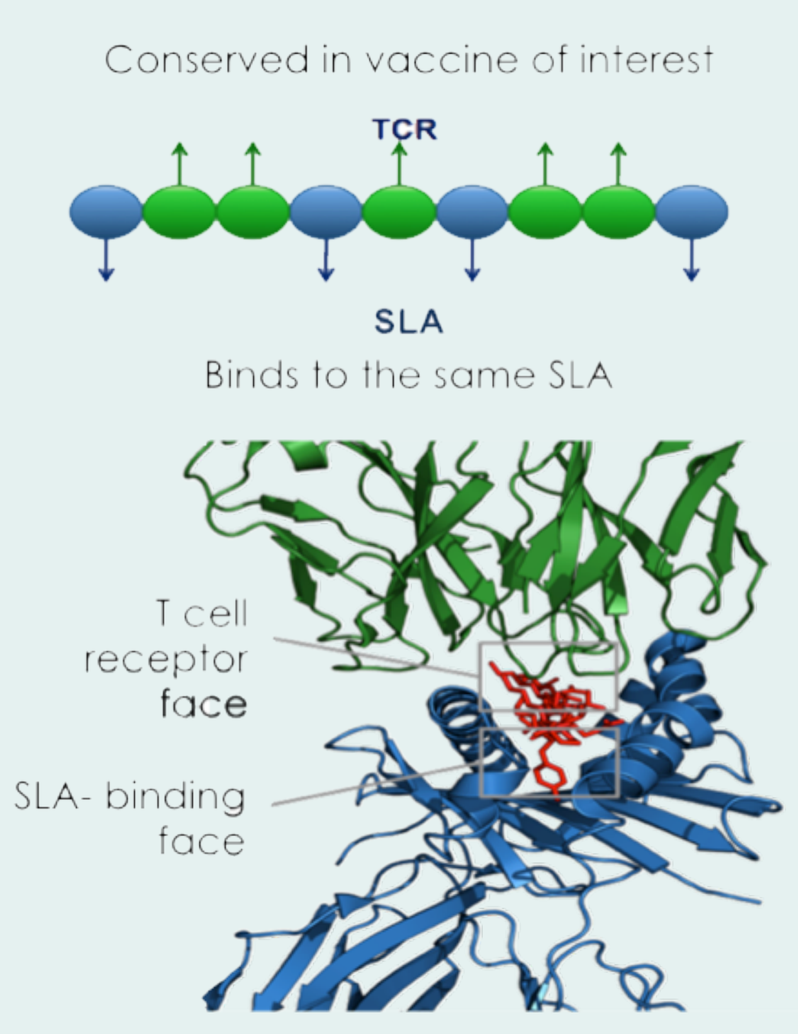
Conserved in vaccine of interest
TCR
SLA
Binds to the same SLA
T cell receptor face
SLA- binding face

T cell epitope content comparison (EpiCC)

- Built based on both PigMatrix and JanusMatrix
- Facilitates pairwise comparison of protein sequences based on T cell epitope content rather than sequence identity
- Capable to classify swine IAV strain relatedness to estimate cross-protective potential of a vaccine strain for circulating viruses

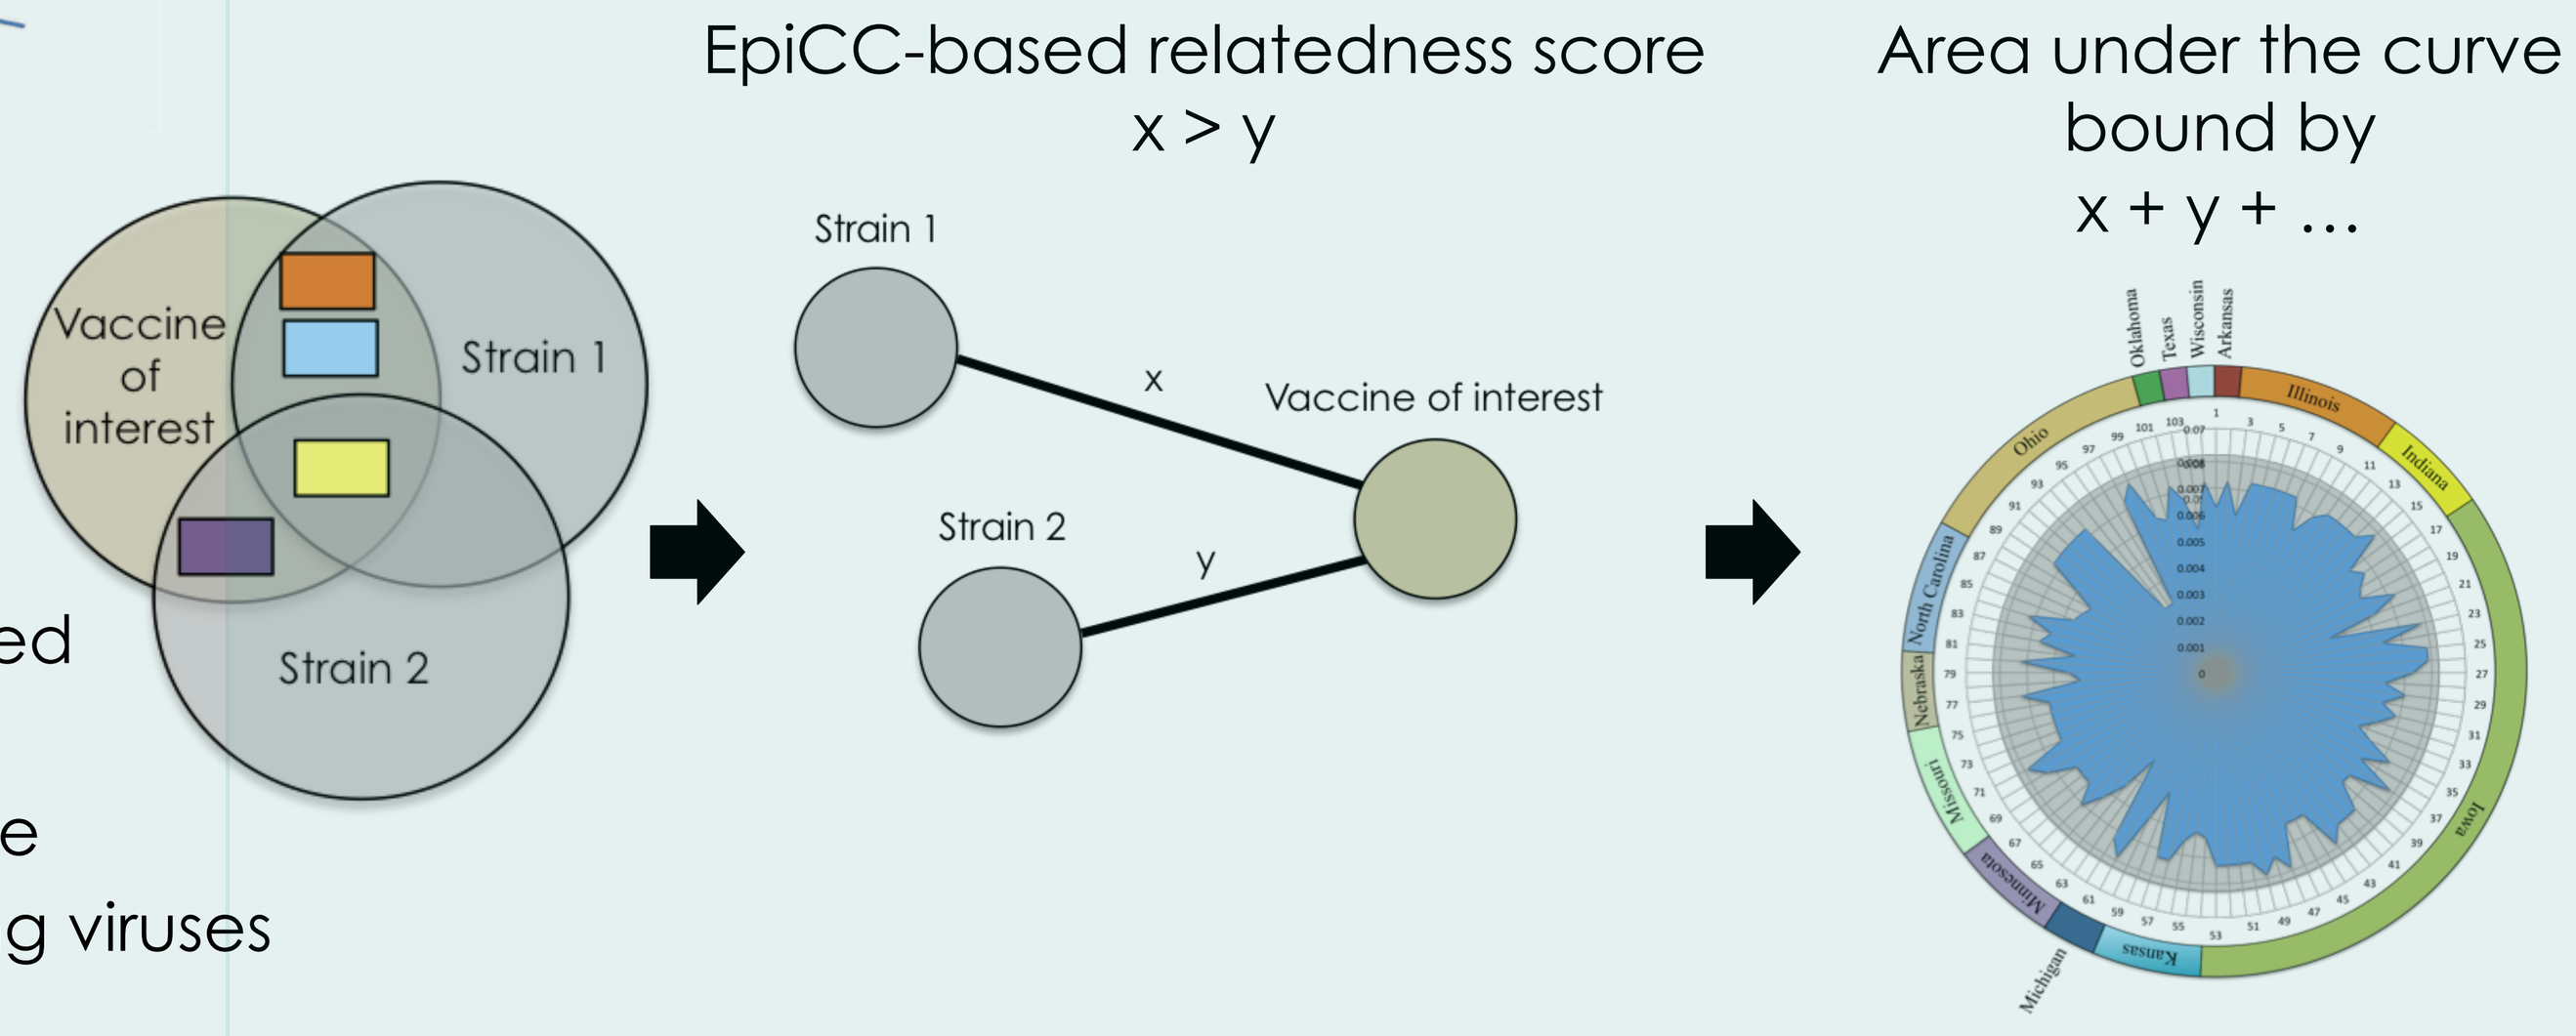
Vaccine of interest
Strain 1
Strain 2
EpiCC-based relatedness score
x > y
Strain 1
x
Vaccine of interest
Strain 2
y
Area under the curve bound by
x + y + …

Figure 3| Relatedness of IAV strains to the DNA epitope-based vaccine

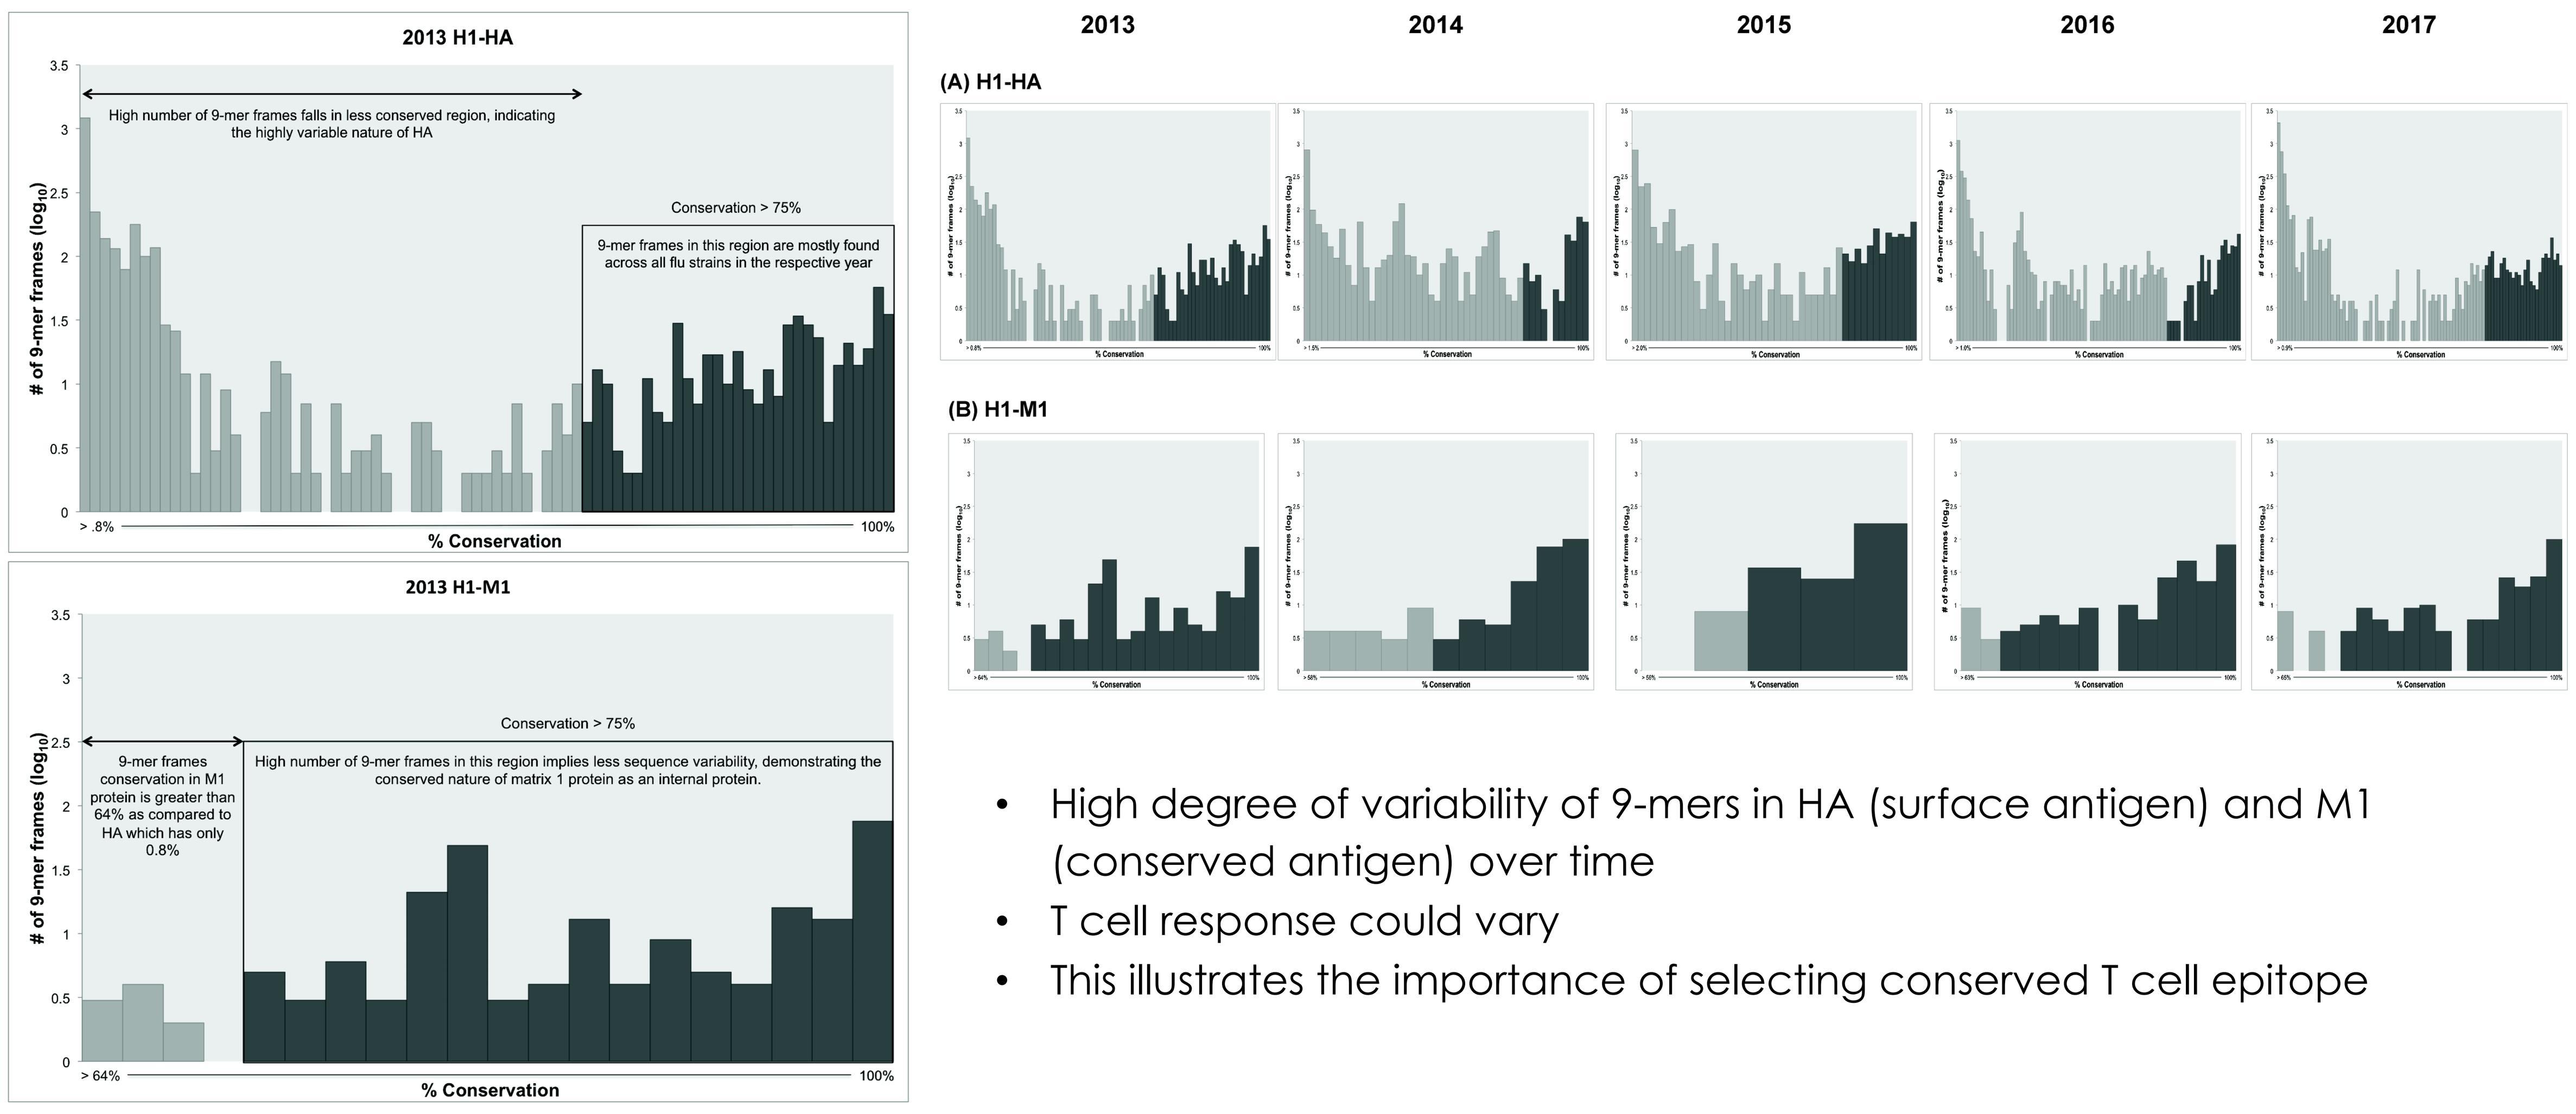
2013 H1-HA
High number of 9-mer frames falls in less conserved region, indicating the highly variable nature of HA
Conservation > 75%
9-mer frames in this region are mostly found across all flu strains in the respective year
of 9-mer frames (log10)
% Conservation
2013 H1-M1
Conservation > 75%
9-mer frames conservation in M1 protein is greater than 64% as compared to HA which has only 0.8%
High number of 9-mer frames in this region implies less sequence variability, demonstrating the conserved nature of matrix 1 protein as an internal protein.
2013
2014
2015
2016
2017
(A) H1-HA
(B) H1-M1

- High degree of variability of 9-mers in HA (surface antigen) and M1 (conserved antigen) over time
- T cell response could vary
- This illustrates the importance of selecting conserved T cell epitope

Figure 4| T cell epitope conservation between the T cell-directed multi-epitope DNA vaccine against swine IAV circulating strains over five years

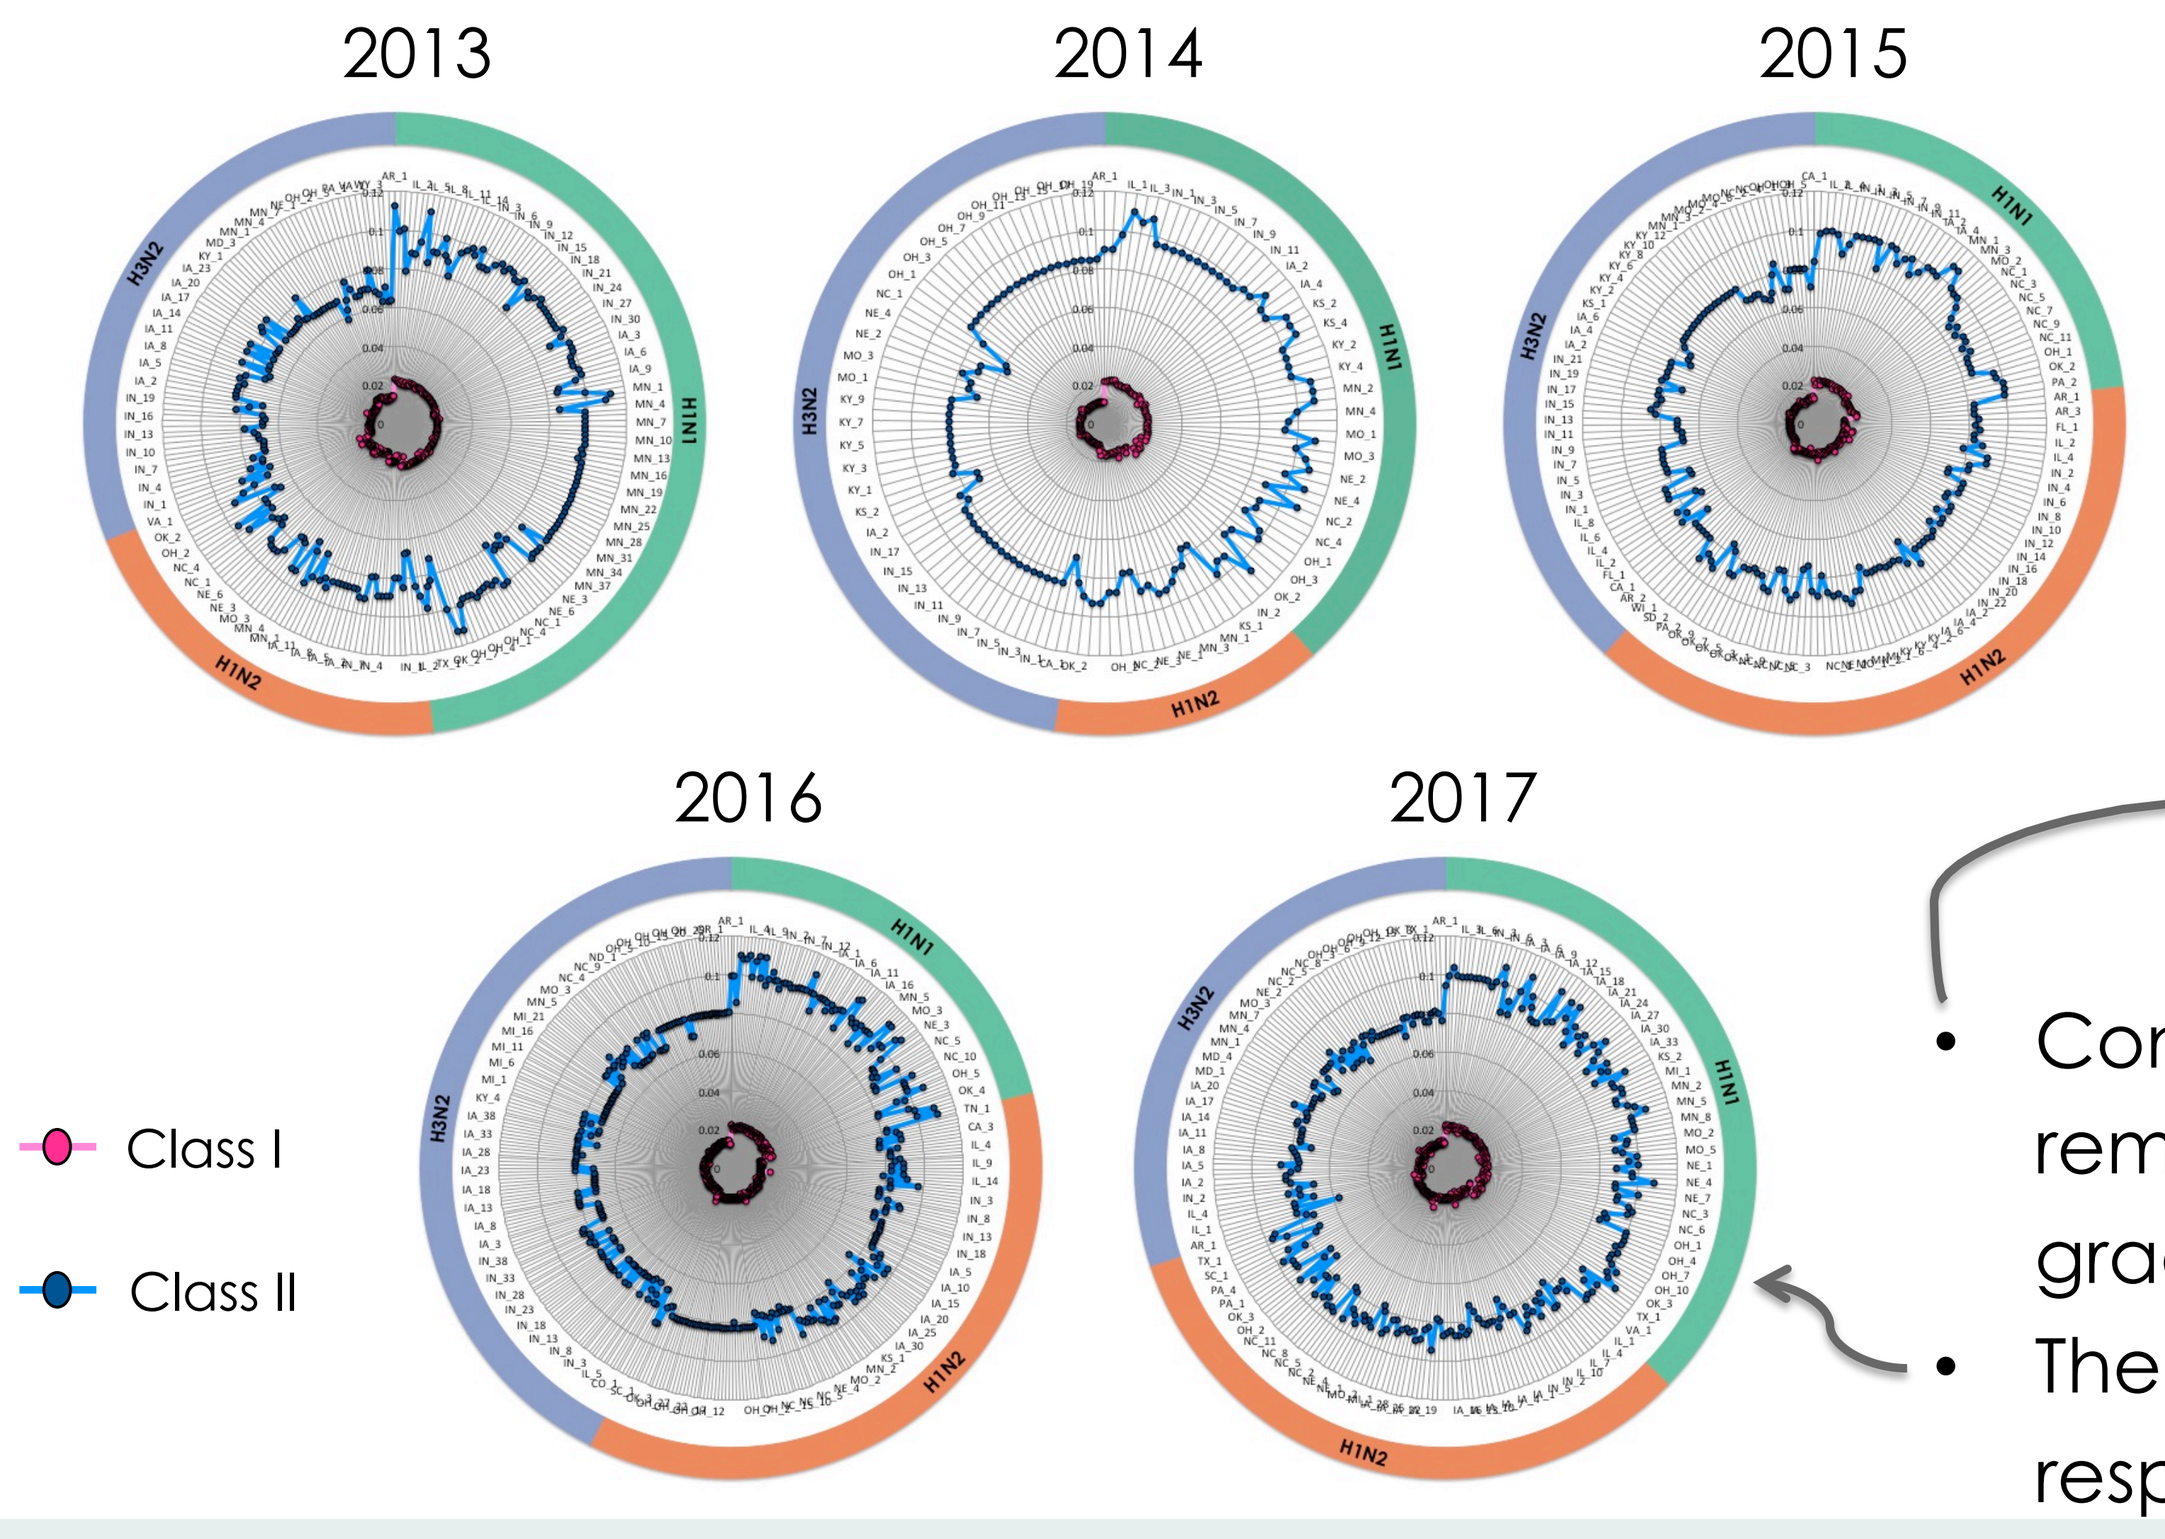
2013
2014
2015
2016
2017
Class I
Class II

Figure 5| T cell epitope content conservation comparison (HA) of the T cell multi-epitope DNA vaccine and conventional vaccines relative to 2013

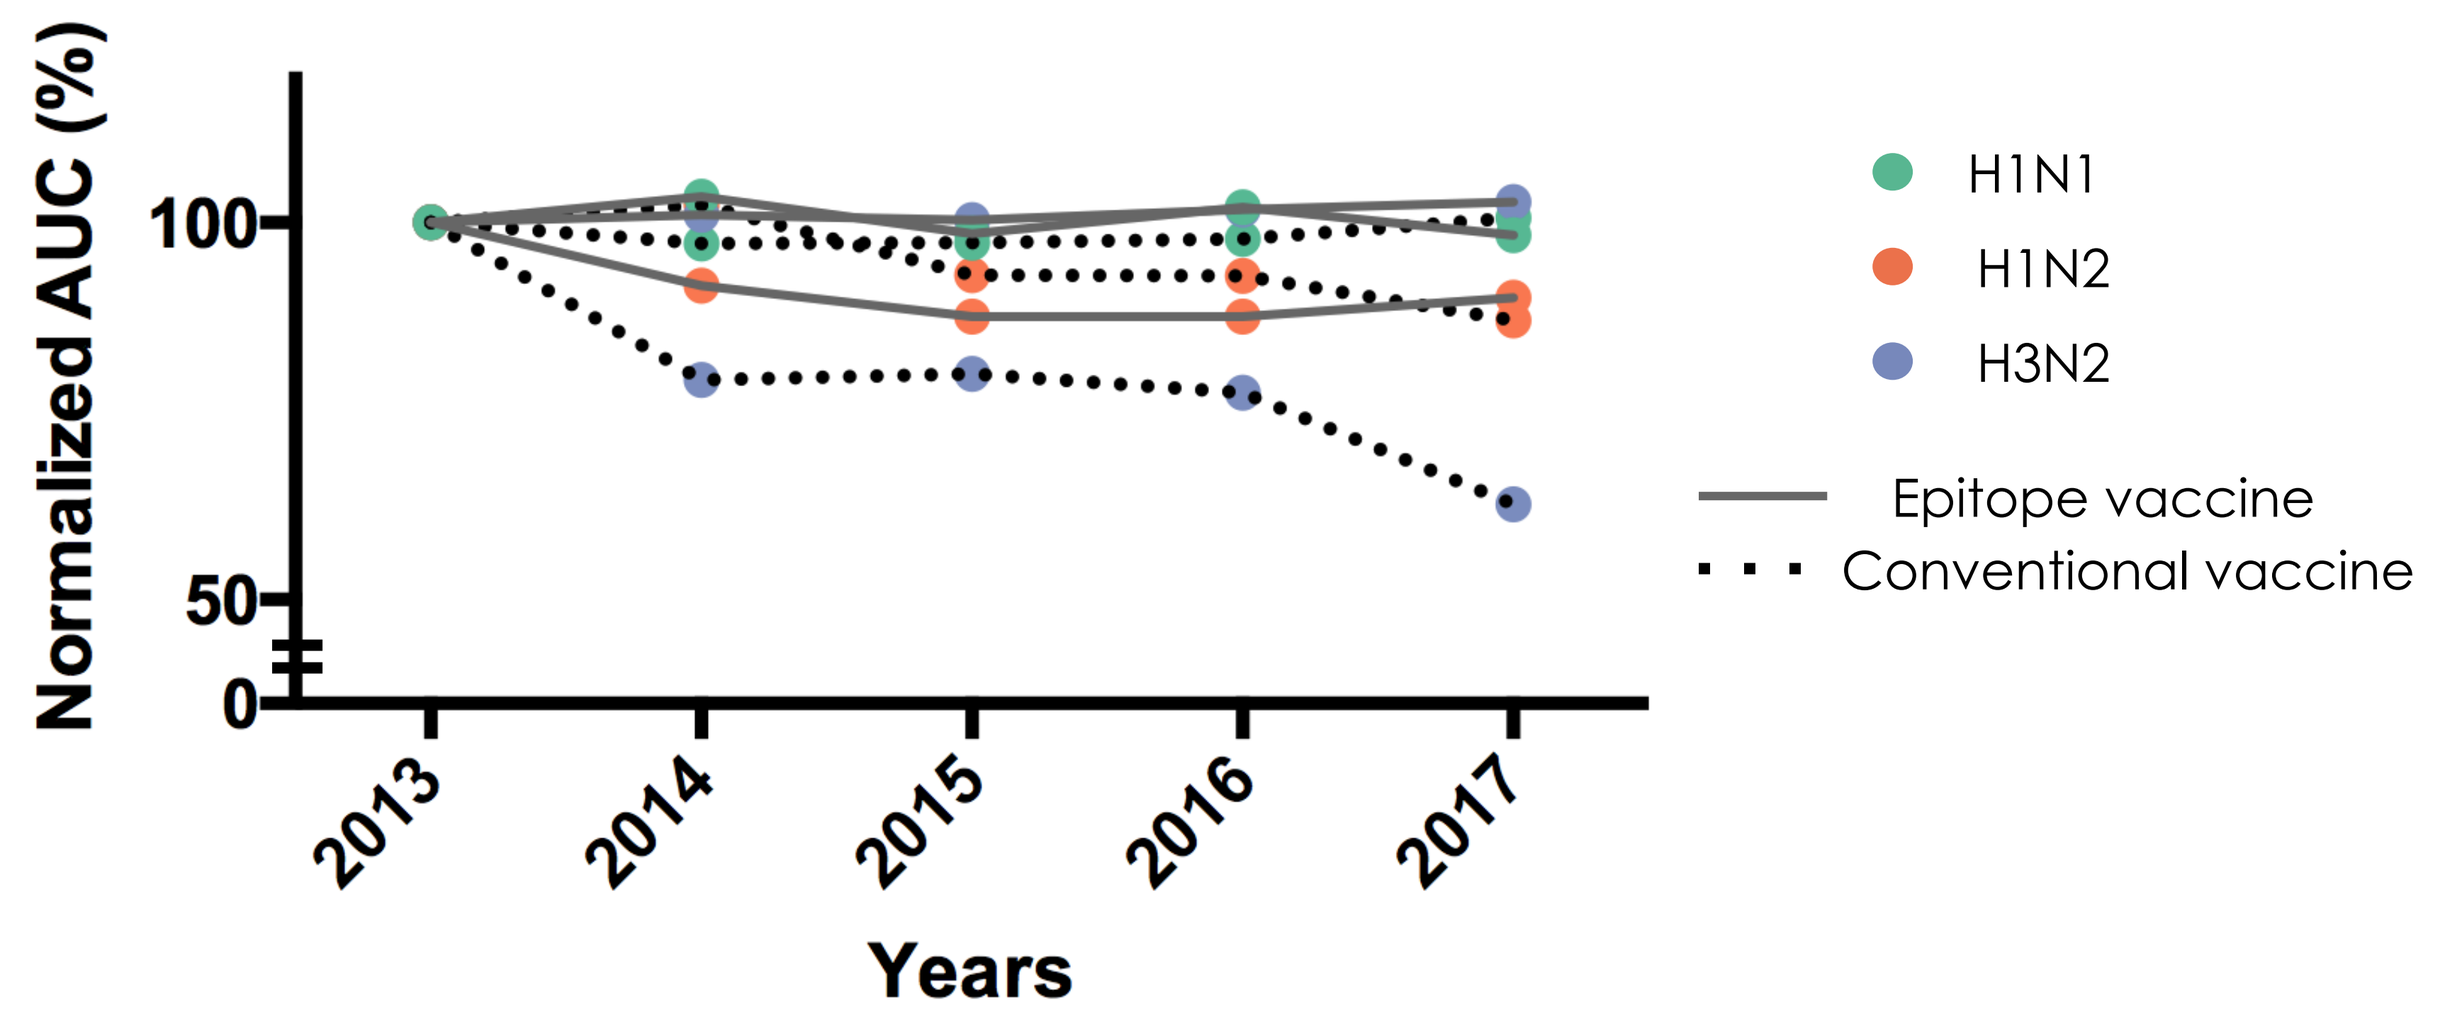
Normalized AUC (%)
100
50
0
2013
2014
2015
2016
2017
Years
H1N1
H1N2
H3N2
Epitope vaccine
Conventional vaccine

- Conservation of T cell epitope content of the multi-epitope DNA vaccine remains high and steady compared to conventional vaccines that gradually reduce over time
- The multi-epitope DNA vaccine is able to drive better CD4 immune response

References:
1) Gutiérrez, A. H., C. Loving, L. Moise, F. E. Terry, S. L. Brockmeier, H. R. Hughes, W. D. Martin, and A. S. De Groot. 2016. In vivo validation of predicted and conserved T cell epitopes in a swine influenza model. PLoS One 11.
2) Gutiérrez, A., Rapp-Gabrielson, V., Terry, F., Loving, C., Moise, L., Martin, W., & De Groot, A. (2017). T-cell epitope content comparison (EpiCC) of swine H1 influenza A virus hemagglutinin. Influenza and Other Respiratory Viruses, 11(6), 531-542.
3) https://www.npr.org/sections/health-shots/2019/06/11/729314248/to-save-the-science-poster-researchers-want-to-kill-it-and-start-over

Contact information: Dr. Anne S. De Groot (dr.annie.degroot@gmail.com)

THE UNIVERSITY OF RHODE ISLAND
THINK BIG WE DO

cubed Institute for Immunology & Informatics

EpiVax

Get poster handout

SCAN ME